312

Redirecting T cells to tumor targets with functionally diverse CD3-binding antibodies

Lindsay DeVorkin, Tim M Jacobs, Raffi Tonikian, Karine Herve, Kate Caldwell, Yuri Hwang, Cristina Faralla, Wei Wei, Katherine J Lam, Harveer Dhupar, Tran NT Tran, Melissa Cid, Lena M Bolten, Tova Pinsky, Juntao (Matt) Mai, Ping Xiang, Courteney Lai, Ahn Lee, Patrick Chan, Jasmine Chin, Sherie Duncan, Aaron Yamniuk, Kush Dalal, Kevin A Heyries, **Bryan C Barnhart**

AbCellera

COPYRIGHT © ABCELLERA

THE CHALLENGE

CD3 T cell engagers bridge the gap between cancer and the immune system by redirecting T cells to tumor targets, regardless of their specificity. But with hundreds of bispecific CD3 T cell engagers in development, there are only two approved molecules on the market.

CD3 T cell engager discovery has been limited because diverse panels of parental antibodies are hard to produce, and the pairing of parental antibodies is hard to perfect.

The intensity of T cell activation is a key predictor of safety and efficacy and is, in part, determined by the affinity and epitope of the CD3-binding arm. CD3-binders that strongly activate T cells can trigger dose-limiting toxicities, including cytokine release syndrome. On the other hand, CD3- binders that weakly activate T cells can lack potency. The result is a narrow therapeutic window for T cell engager development.

In addition, a unique combination of CD3- and tumor-binding arms are needed for each cancer type to allow for the binding of two targets, at the same time, with the right 3D geometry.

THE SOLUTION

Begin with diverse panels of parental antibodies to increase the probability of finding lead bispecific candidates that are effective and scalable, reducing the need for downstream engineering to eliminate liabilities.

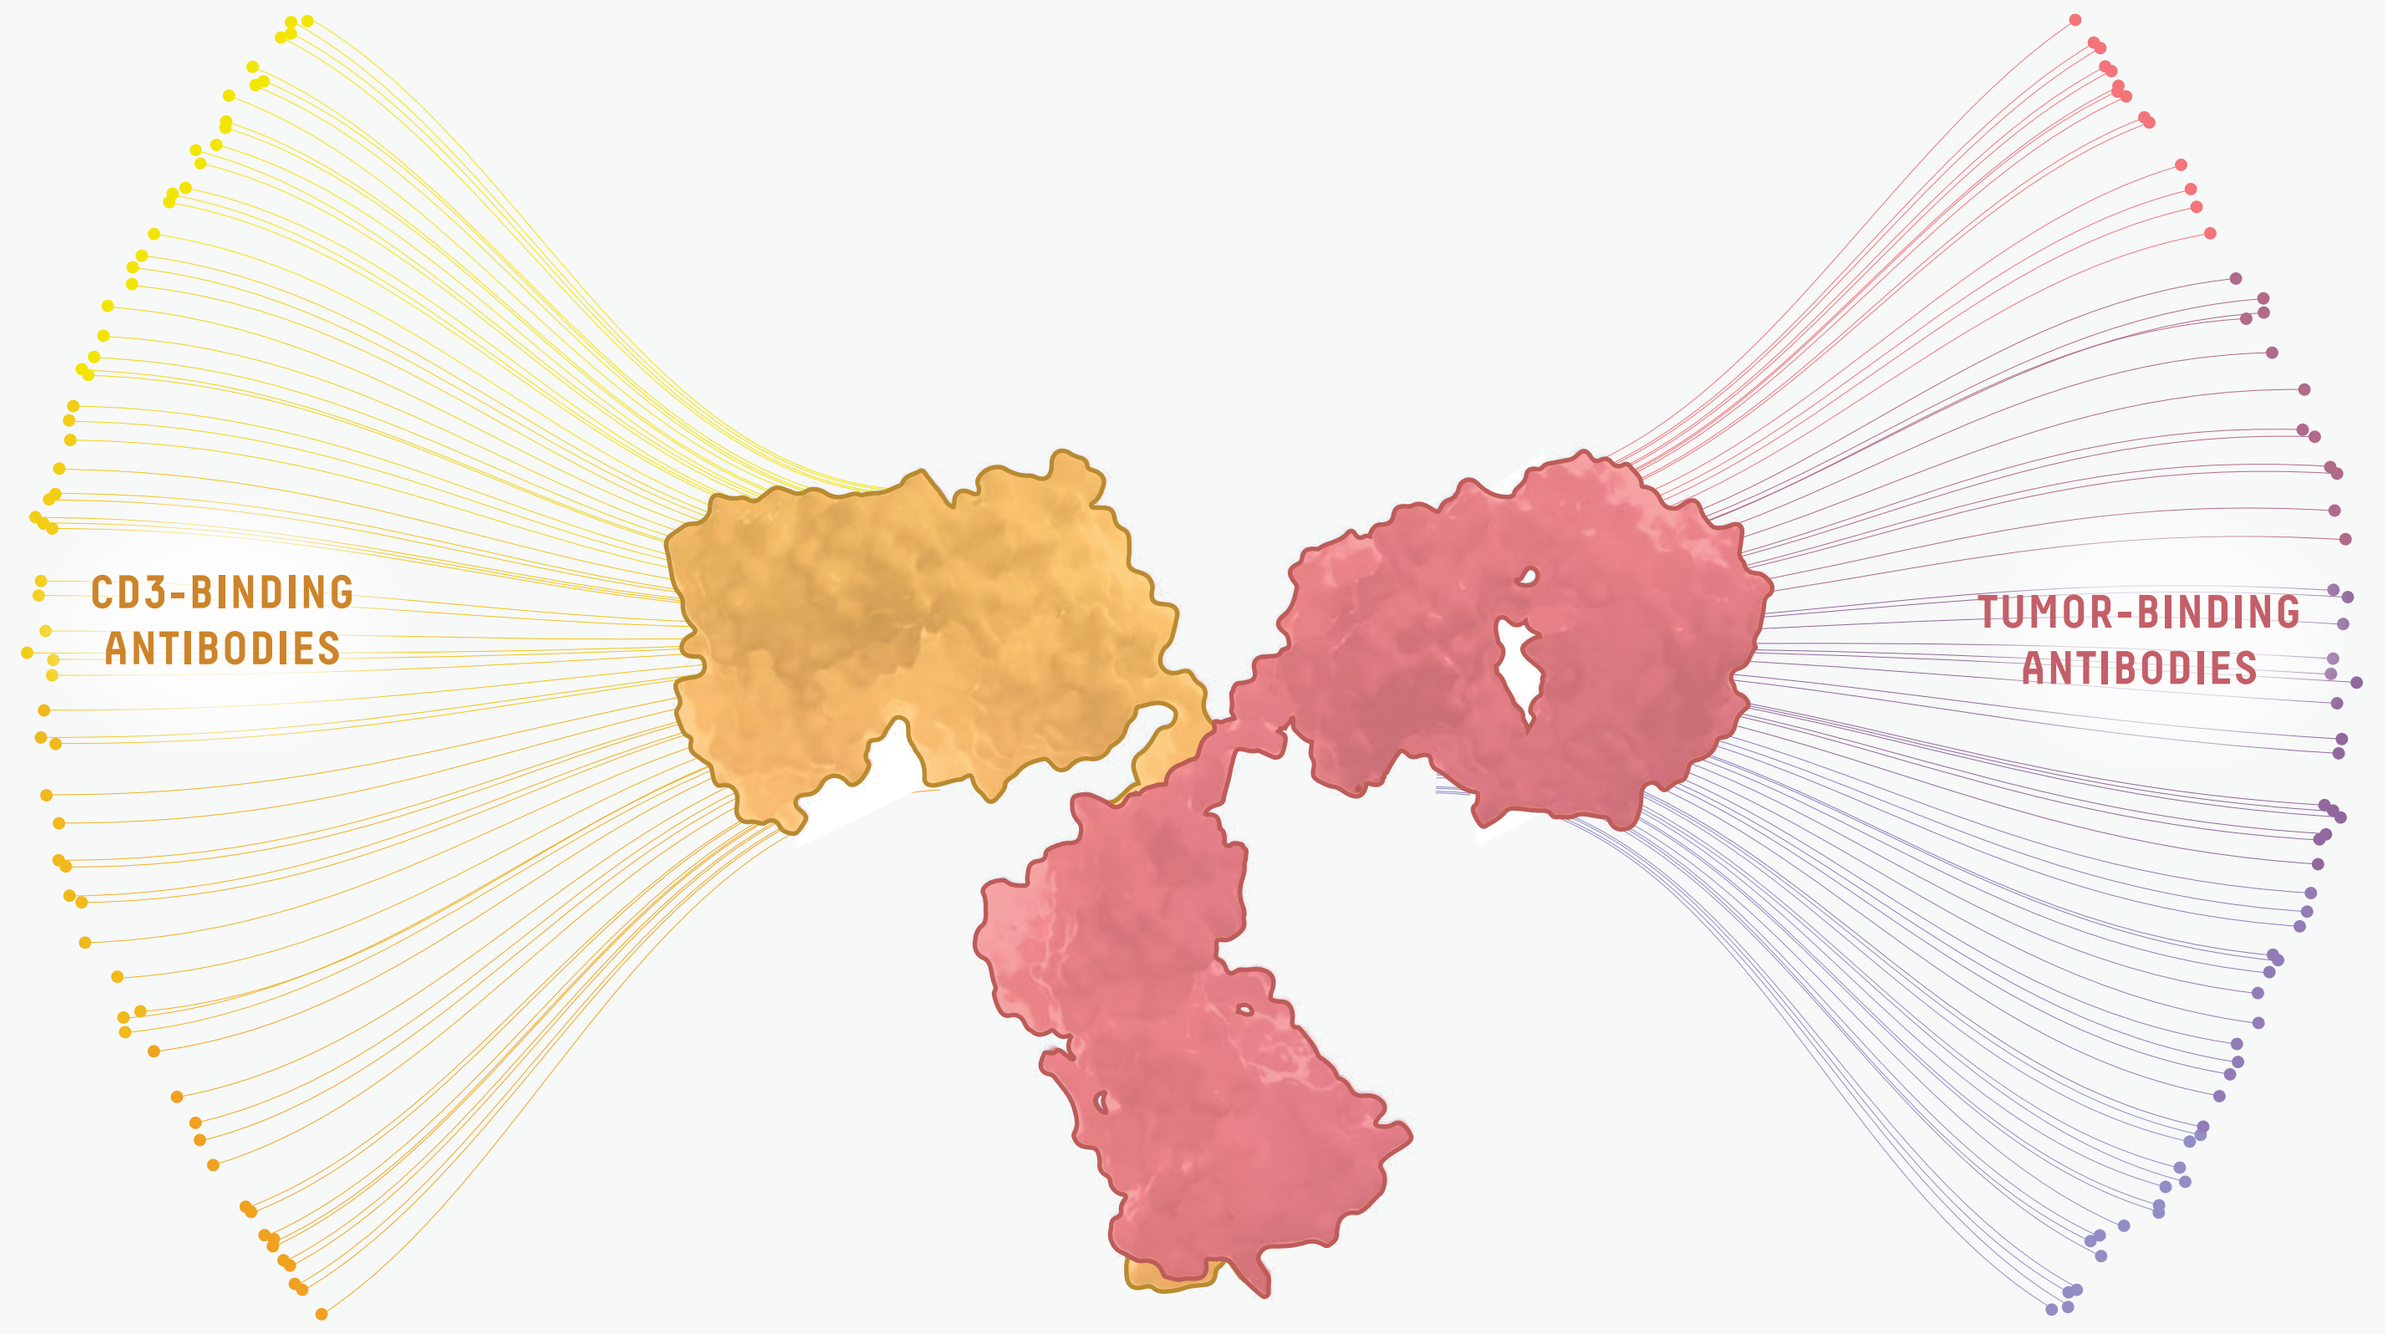

CD3-BINDING ANTIBODIES
TUMOR-BINDING ANTIBODIES

AbCellera's panel of fully human CD3-binding antibodies are diverse, developable, and validated.

Our integrated technology stack discovered CD3-binding antibodies that:

- include **human/cyno cross-reactive binders**
- bind CD3 with a **range of affinities**
- activate T cells with a **range of potencies**
- have **favorable developability properties**

Our clinically-validated bispecifics platform, OrthoMab™, generated CD3 binders that:

- **activate T cells** in bispecific formats
- **result in tumor cell killing** with a range of potencies

DIVERSE

We identified diverse antibody sequences, optimized by nature.

Proprietary immunization strategies using humanized mice enriched antibody diversity from the start.

Bioinformatic analysis revealed diverse antibody sequences with a range of V genes and CDR3 lengths, optimized by natural selection through somatic hypermutation.

3.5 million cells screened
1,653 CD3-specific hits
275 unique antibodies
81 clonal families

Maturation

Identity to Germline

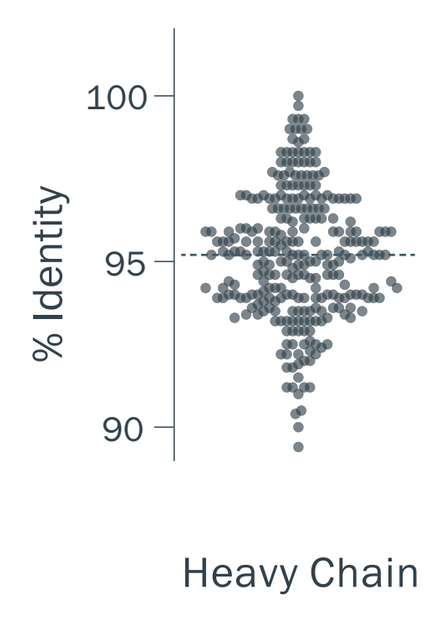

% Identity
100
95
90
Heavy Chain

Sequence Diversity

CDR3 Length

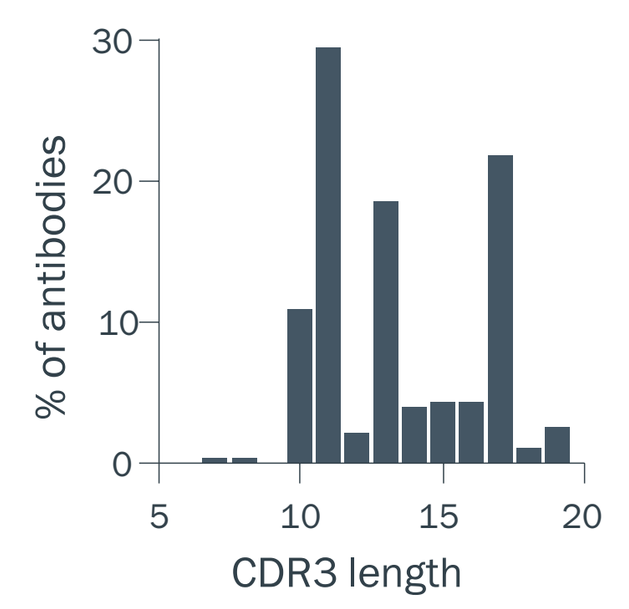

% of antibodies
CDR3 length

V Gene Usage

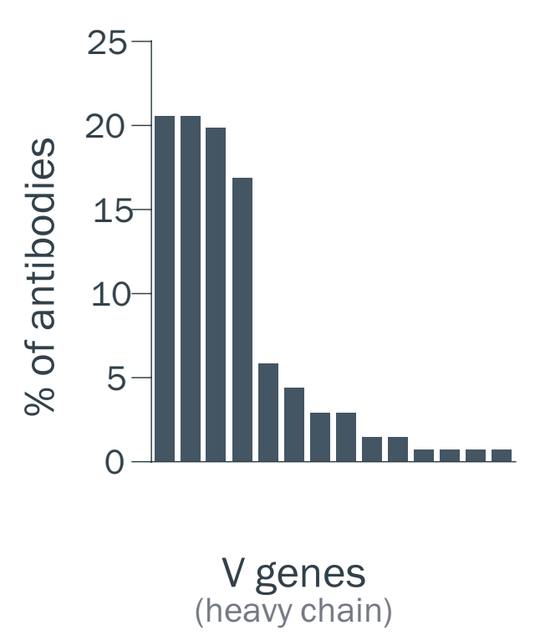

% of antibodies
V genes
(heavy chain)

We found functionally diverse CD3 binders that strike the right balance.

High-throughput antibody expression identified functionally diverse CD3-specific antibodies, including human + cyno cross-reactive binders.

Antibodies bound human CD3 with a **broad range of affinities** (K_D values from ~1 nM to 1 µM).

Antibodies were tested for T cell activation by CD69 and CD25 upregulation.

All antibodies tested activated T cells with a **broad range of potencies** (EC_{50} values from ~6 nM to 200 nM).

CD3 Affinity

Binding Affinity to Human CD3

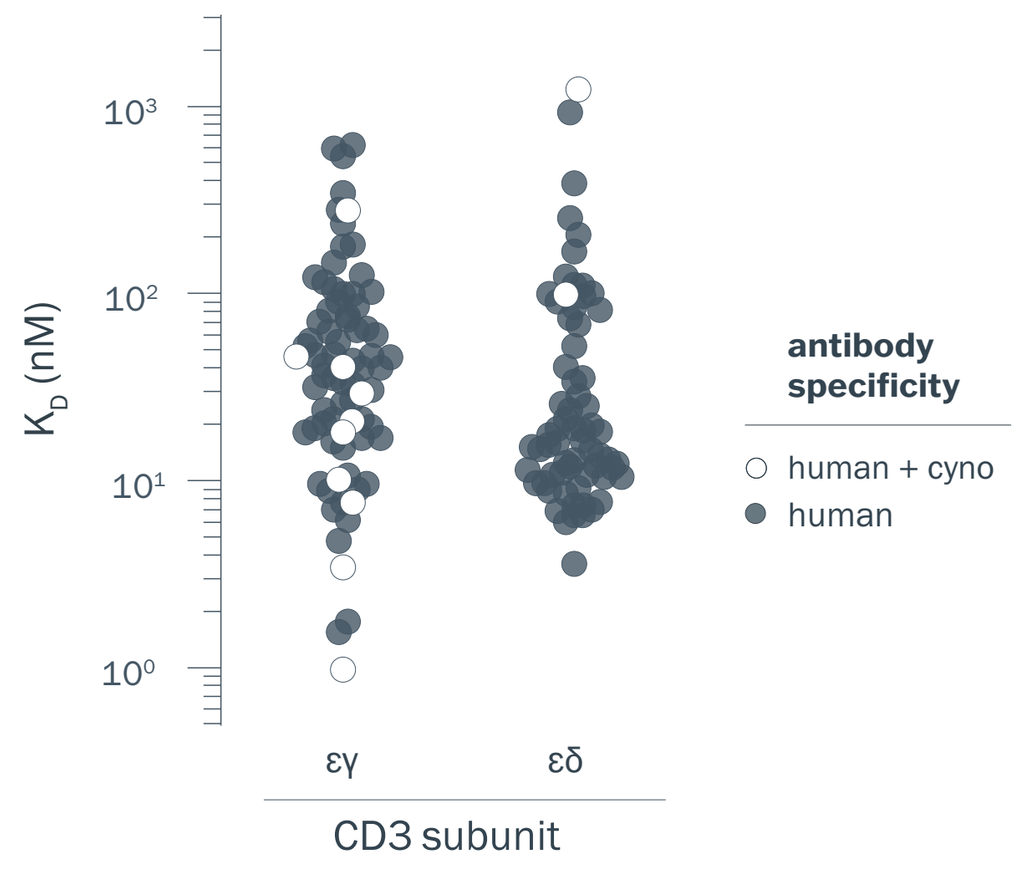

KD (nM)
εγ
εδ
CD3 subunit
antibody specificity
human + cyno
human

T Cell Activation

CD69 and CD25 Upregulation

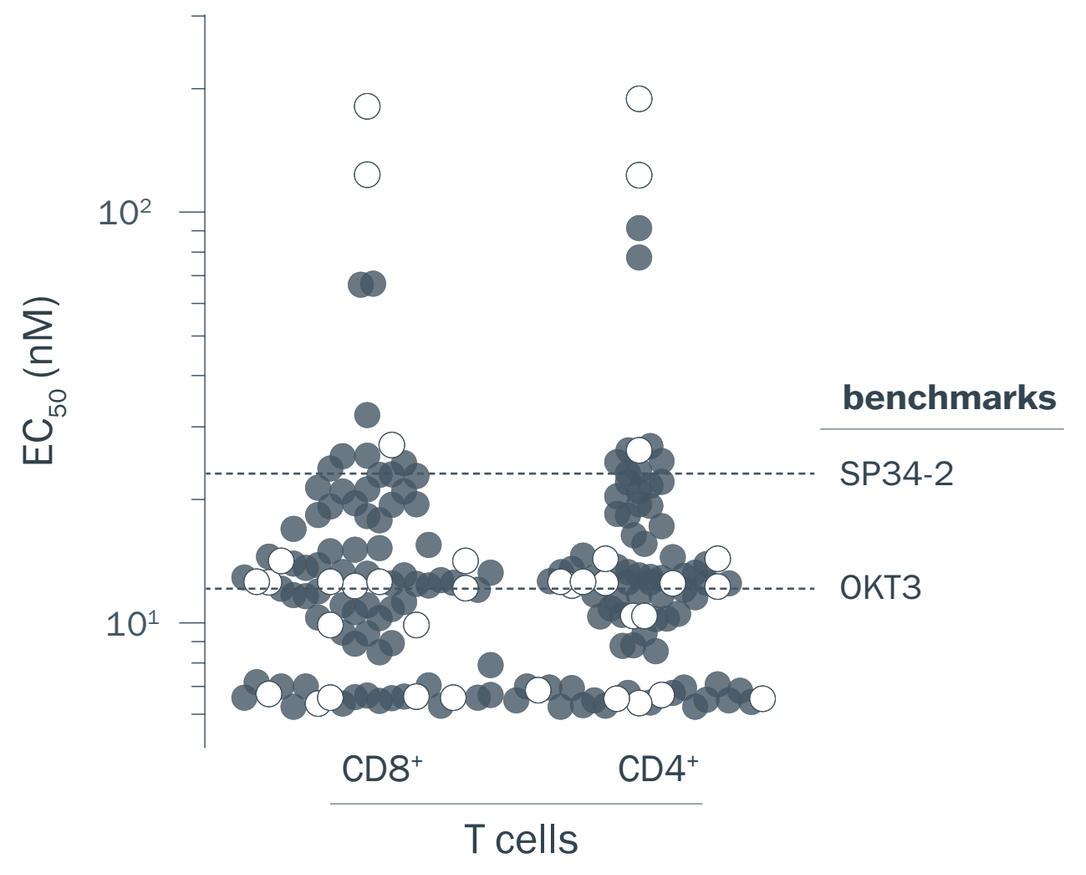

EC50 (nM)
benchmarks
SP34-2
OKT3
CD8+
CD4+
T cells

DEVELOPABLE

We selected scalable antibodies with favorable biophysical properties.

High-throughput antibody assessment identified antibodies with promising developability traits:

Low mean hydrophobicity by high-throughput analytical hydrophobic interaction chromatography (aHIC)

Low mean self-association by affinity-capture self-interaction nanoparticle spectroscopy (AC-SINS)

Low mean polyspecificity by baculovirus particle (BVP)-ELISA

Hydrophobicity

aHIC

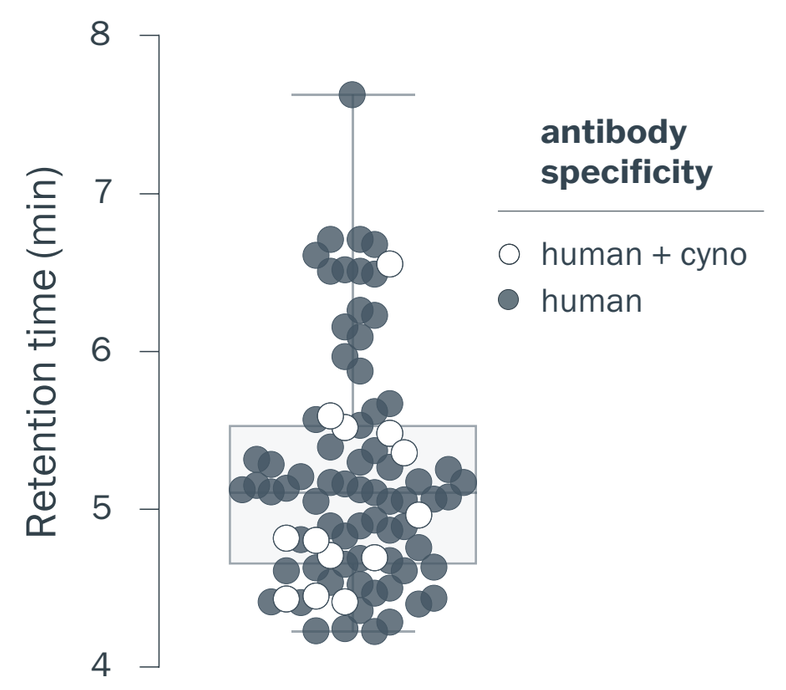

Retention time (min)
antibody specificity
human + cyno
human

Self-Association

AC-SINS

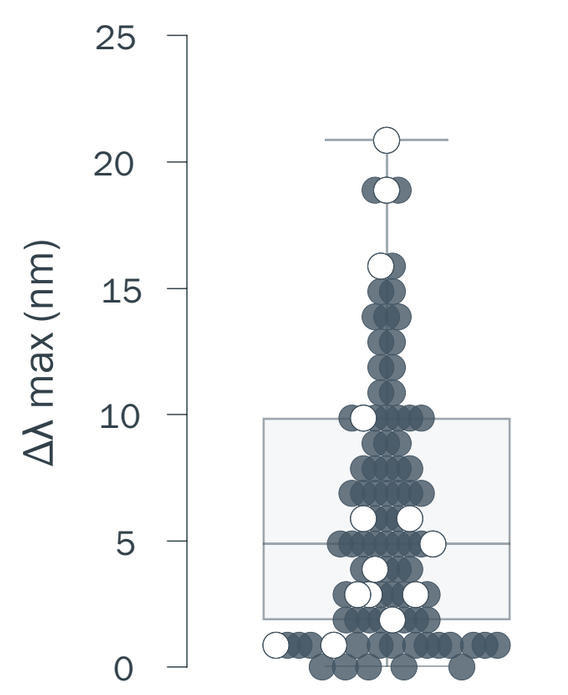

Δλ max (nm)

Polyspecificity

BVP-ELISA

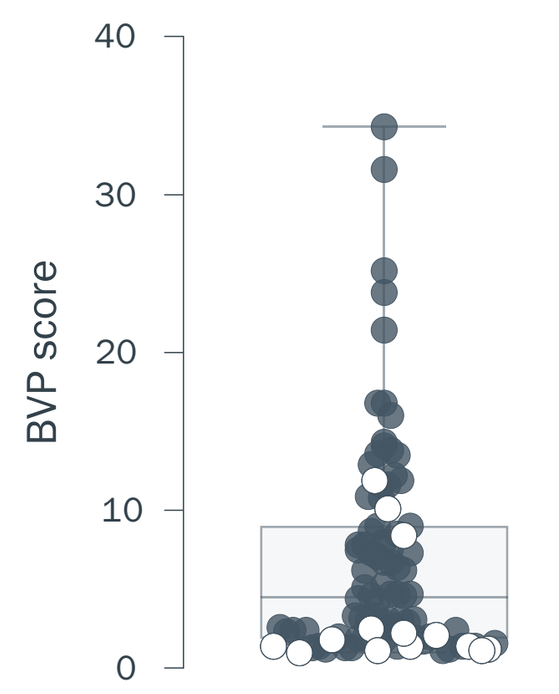

BVP score

VALIDATED

We validated CD3 binders in bispecific formats.

19 CD3 binders were tested in CD3 x EGFR bispecific formats using our clinically-validated bispecifics platform, OrthoMab™.

95% of the bispecific antibodies activated T cells in an NFAT reporter assay with isogenic CHO-K1 $EGFR^{low}$ and $EGFR^{med}$-expressing cell lines.

A single EGFR-binding arm was used for validation, and benchmark CD3 x EGFR bispecific antibodies were generated from clinical-stage monoclonal antibodies.

Bispecific Engineering

Powered by OrthMab™

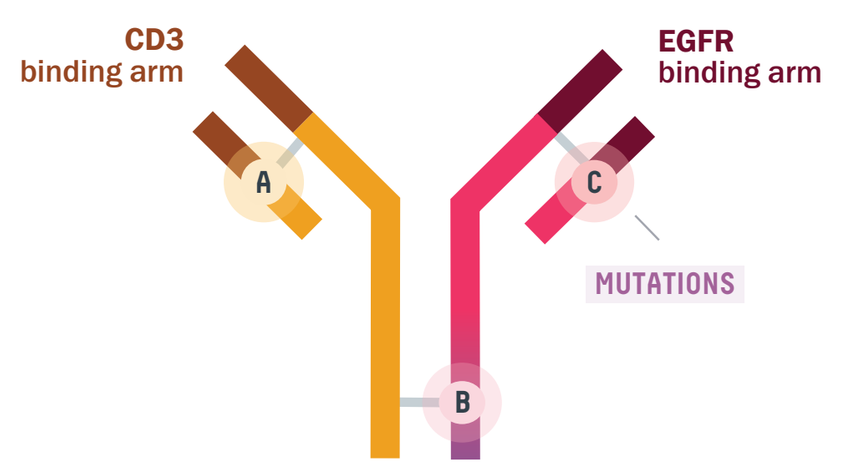

CD3 binding arm
EGFR binding arm
A
B
C
MUTATIONS

3 protein-protein interfaces with 1 to 4 mutations per domain

T Cell Activation

NFAT reporter assay

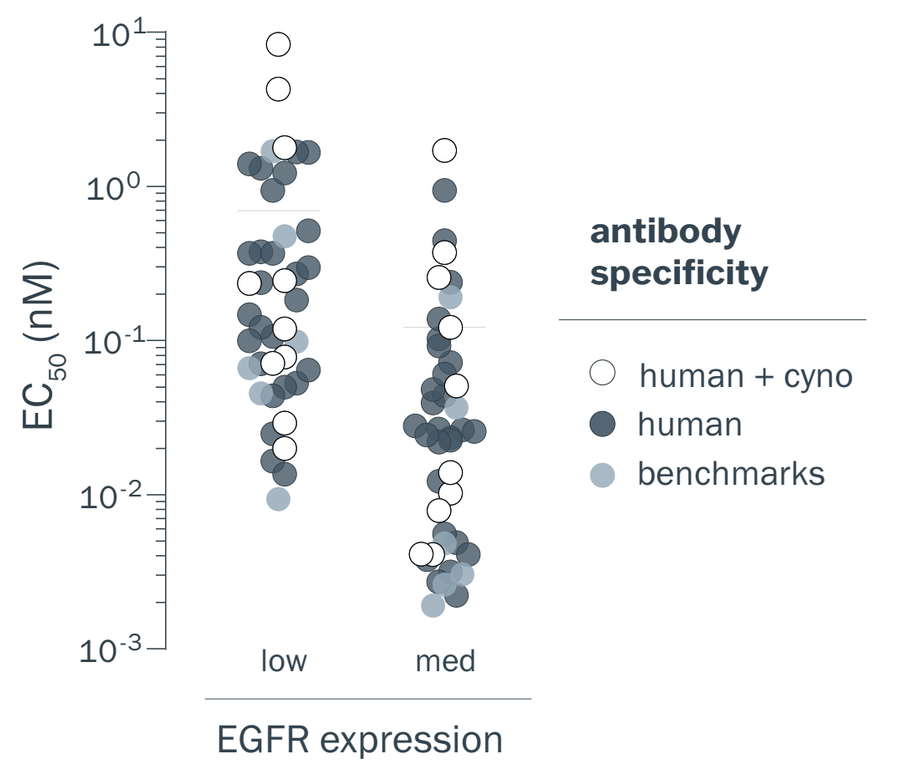

EC50 (nM)
low
med
EGFR expression
antibody specificity
human + cyno
human
benchmarks

Cell Lines	EC_{50} Values
$EGFR^{low}$	~10 pM to 8 nM
$EGFR^{med}$	~2 pM to 2 nM

We translated diversity into promising bispecific antibodies.

Integration of our diverse panel of CD3-binding antibodies with OrthoMab™ and antibody analytics enabled identification of CD3 T cell engagers that we believe have the highest probability of success.

CD3 x EGFR bispecific molecules 1 and 2 showed **potent T cell activation** in an NFAT reporter assay, and **tumor cell killing** in an xCELLigence assay.

Both were derived from CD3 parental antibodies with **human + cyno cross-reactivity**, and parental arms were **paired with 99% and 87% efficiencies**, respectively.

PROOF OF CONCEPT Profiles of promising **bispecific antibodies:**

Bispecific Tumor Cell Killing

Molecule 1 Molecule 2

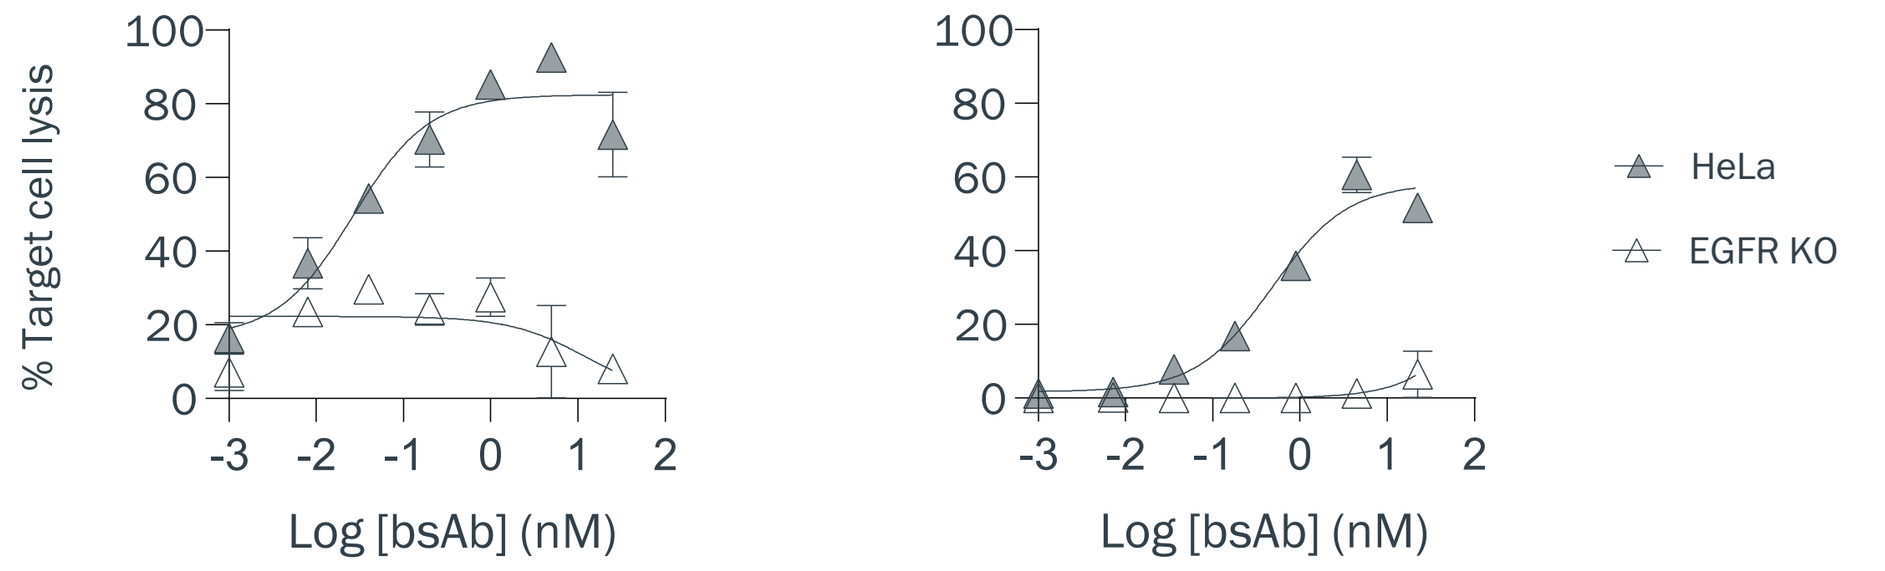

% Target cell lysis
Log [bsAb] (nM)
HeLa
EGFR KO

Bispecific T Cell Activation

Molecule 1 Molecule 2

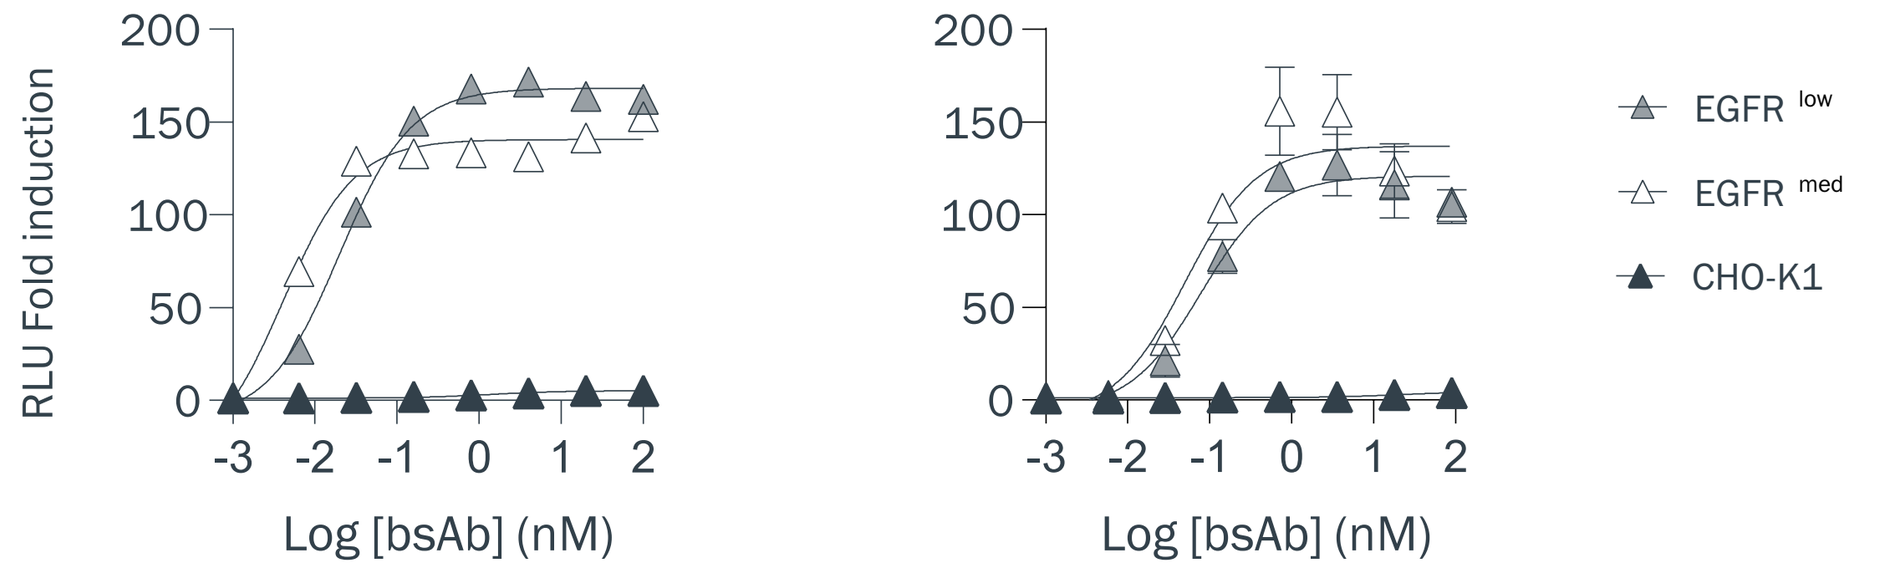

RLU Fold induction
Log [bsAb] (nM)
EGFR low
EGFR med
CHO-K1

CD3 x EGFR Bispecific Antibody

Bispecific Molecule	1	2
Pairing efficiency	99%	87%
T cell activation ($EGFR^{med}$ EC_{50})	0.004 nM	0.05 nM
Tumor cell killing (48 hours EC_{50})	0.026 nM	0.53 nM

CD3 Monoclonal Antibody

Parental Molecule		1	2
Apparent affinity	(K_D) (εδ)	18.4 nM	24.1 nM
T cell activation	($CD8^+$ EC_{50})	12.3 nM	123.4 nM
Hydrophobicity	(retention time)	5.4 min	4.8 min
Self association	(Δγ max)	0.88 nm	2.88 nm
Polyspecificity	(BVP score)	1.8	8.4

DOWNLOAD POSTER