Diverse CD28-binding antibodies for costimulatory T-cell engager development

AUTHORS
Katherine Lam, Shirley Zhi, Lucas Kraft, Elena Vigano, Wei Wei, Cristina Faralla, Esther Odekunle, Melissa Cid, Stefi Carrara, Creagh Briercliffe, Kelly Bullock, Ingrid Knarston, Gesa Volkers, Michael Kennedy, Grace Leung, Bryan C. Barnhart, **Lindsay DeVorkin***, Sherie Duncan
****presenter***

AUTHOR AFFILIATION
AbCellera, Vancouver, Canada

DOWNLOAD POSTER

1859

BACKGROUND

Optimal T-cell activation in the tumor microenvironment requires costimulatory signals

The anti-tumor activity of T cells, initiated via the T-cell receptor (TCR), can be enhanced through costimulatory engagement of the CD28 co-receptor.[1] This costimulation can be leveraged therapeutically using CD28-binding antibodies to improve the efficacy of T-cell activating strategies, such as CD3 T-cell engagers (TCEs), in the tumor microenvironment (Fig. 1).[2,3]

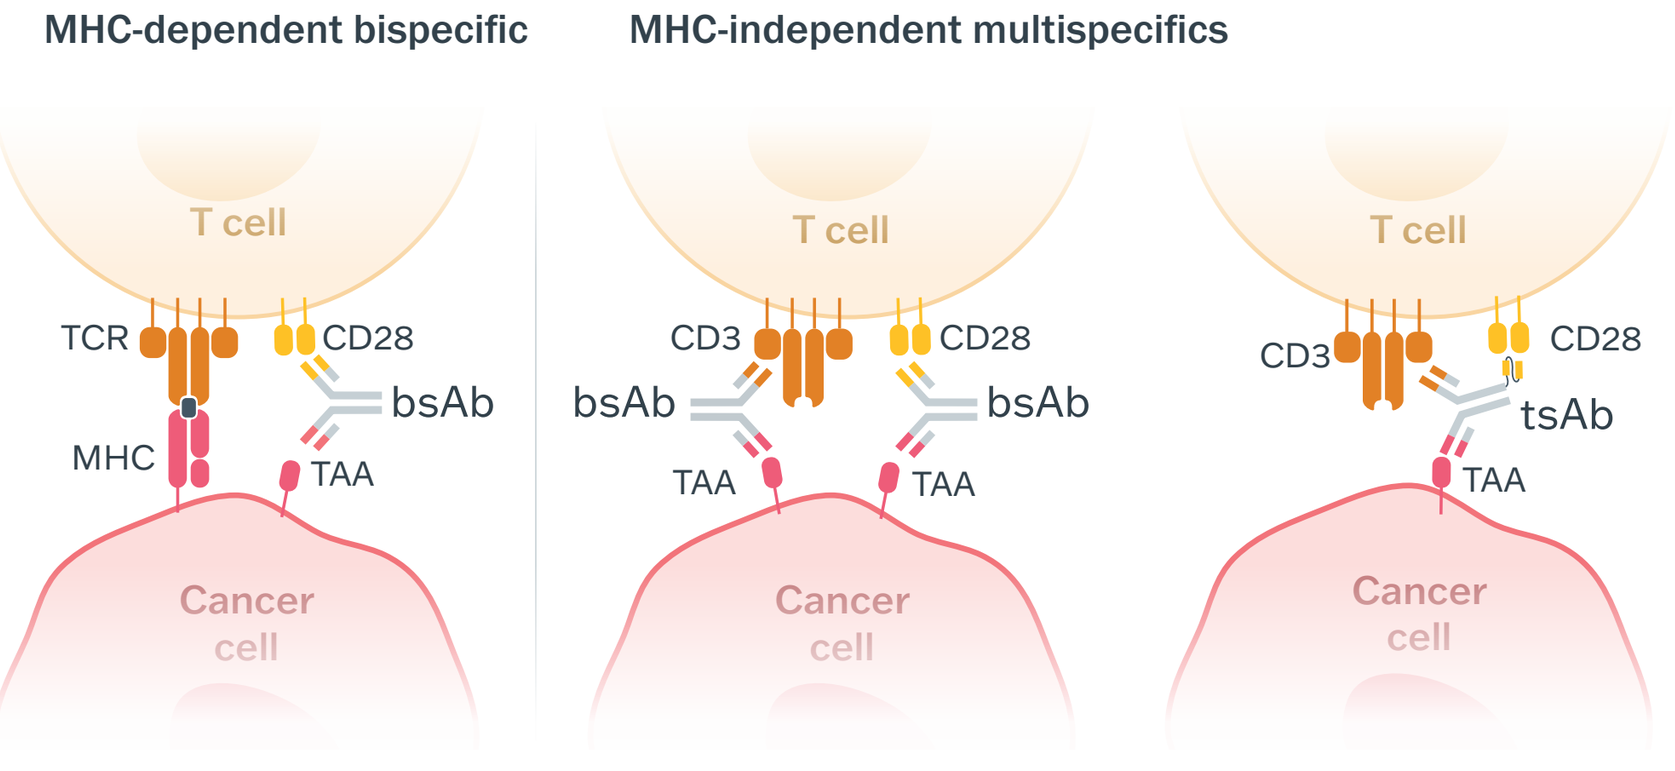
Costimulatory strategies
MHC-dependent bispecific
MHC-independent multispecifics
T cell
TCR
CD28
bsAb
MHC
TAA
CD3
tsAb
Cancer cell

Figure 1. CD28-binding antibodies can enhance anti-tumor activity in combination with T-cell activating therapeutics.

AIM

Generate costimulatory molecules with potential for improved safety and efficacy profiles

CD28-costimulatory antibodies are a promising class of immunotherapies for diverse tumor targets, yet clinical challenges remain.[4,5]

To address these challenges, our aim was to develop a diverse set of CD28-binding IgG and heavy-chain only antibodies (HCAbs) that:

- do not show superagonist activity — a property associated with CD28-mediated toxicities
- have diverse binding properties for the development of immunotherapies across tumor targets and modalities

OUTCOME

Functionally diverse CD28-binding antibodies for T-cell engager development

We generated a panel of CD28-binding HCAbs and IgG antibodies. These antibodies bind a broad range of epitopes, leading to diverse functional activity that is clearly differentiated from benchmarks with known toxicity issues.

Combining these molecules with other T-cell activating strategies represents an opportunity to fine-tune novel costimulatory TCEs for diverse tumor targets.

REFERENCES

1. Huppa JB, Davis MM. (2003). *Nat Rev Immunol*, 3(12):973-83. doi: 10.1038/nri1245.
2. Waite JC, et al. (2020). *Sci Transl Med*, 12(549):eaba2325. doi: 10.1126/scitranslmed.aba2325.
3. Skokos D, et al. (2020). *Sci Transl Med*, 12(525):eaaw7888. doi: 10.1126/scitranslmed.aaw7888.
4. Stein M, et al. (2023). *J of Clin Oncol*, 41:6_suppl. doi: 10.1200/JCO.2023.41.6_suppl.154.
5. Suntharalingam G, et al. (2006). *N Engl J Med*, 355(10):1018-28. doi: 10.1056/NEJMoa063842.
6. Murphy et al. (2019). *Patent Application Publication*, Pub. No: US 2019/0389951 A1.
7. Findlay L, et al. (2010). *J Immunol Methods*, 352(1-2):1-12. doi: 10.1016/j.jim.2009.10.013.

COPYRIGHT © ABCELLERA

CD28-binding antibodies enhance T-cell activity without superagonism

Broad range of costimulatory T-cell activity with differentiation from clinical benchmarks

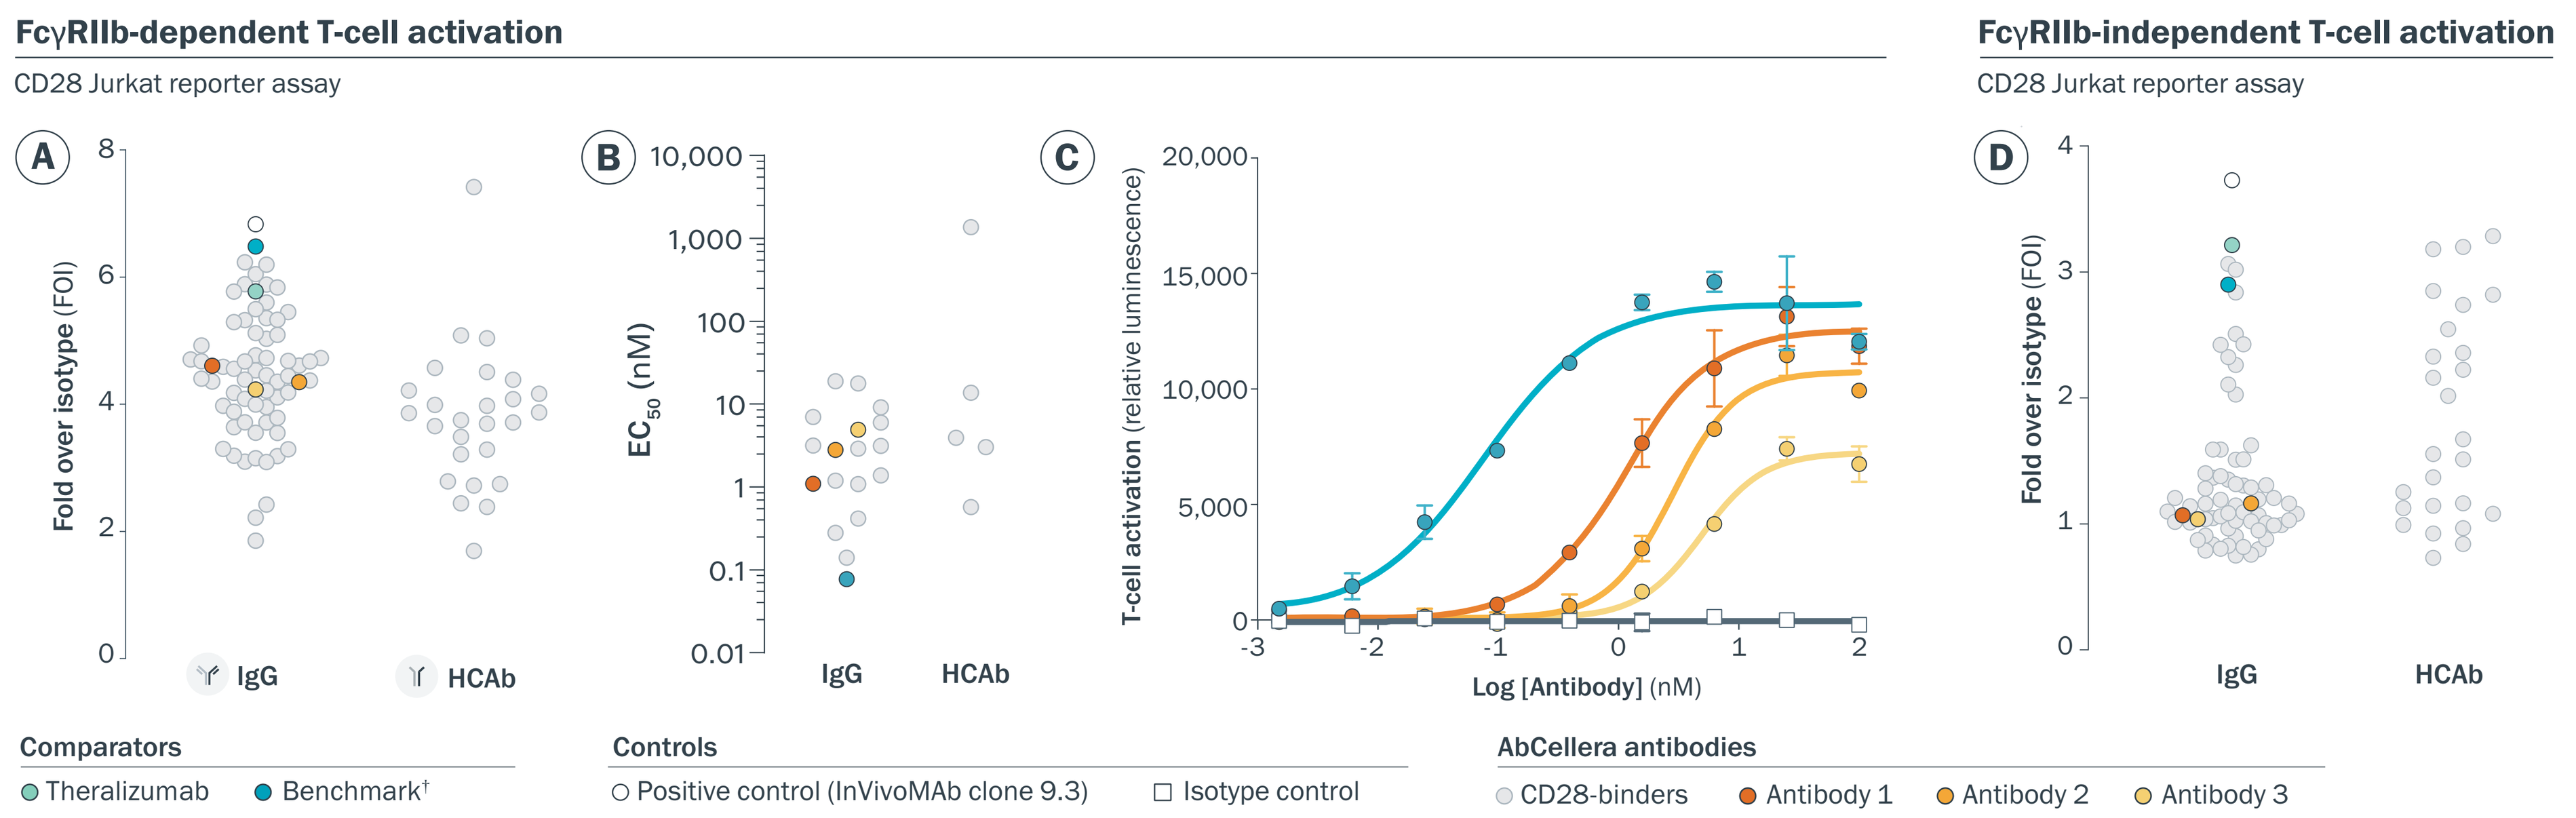
FcγRIIb-dependent T-cell activation
CD28 Jurkat reporter assay
FcγRIIb-independent T-cell activation
CD28 Jurkat reporter assay
Fold over isotype (FOI)
EC50 (nM)
T-cell activation (relative luminescence)
Log [Antibody] (nM)
IgG
HCAb
Comparators
Theralizumab
Benchmark†
Controls
Positive control (InVivoMAb clone 9.3)
Isotype control
AbCellera antibodies
CD28-binders
Antibody 1
Antibody 2
Antibody 3

Figure 2. CD28 activation in the presence or absence of FcγRIIb confirms that antibodies are conditional CD28 agonists, with the majority showing lower crosslinking-independent activation of effector cells than superagonist benchmarks.
(A) 92 antibodies, generated with human IgG1 Fc, were tested for CD28 FcγRIIb-dependent activation. Antibodies were incubated with effector cells endogenously expressing TCR, CD3, and CD28, and CHO-K1 cells engineered to express FcγRIIb and a TCR-engaging protein (Promega JA9331). Relative luminescence for each antibody was normalized to the isotype control (fold over isotype ⩾ 1 considered positive). Three example antibodies (1-3) with diverse functional responses are highlighted. **(B)** 22 antibodies were selected for a 1-in-4, nine-point titration series starting at 100 nM, EC_{50} values are shown. **(C)** Relative luminescence for example antibodies, benchmark, and a human IgG isotype control are shown. **(D)** FcγRIIb-independent agonist activities were similarly assessed using CHO-K1 cells engineered to express a TCR-engaging protein, but not FcγRIIb. †*Bivalent monoclonal CD28 benchmark antibody generated using sequences from REGN5678 patent.*[6]

CD28-binding antibodies do not show superagonist activity

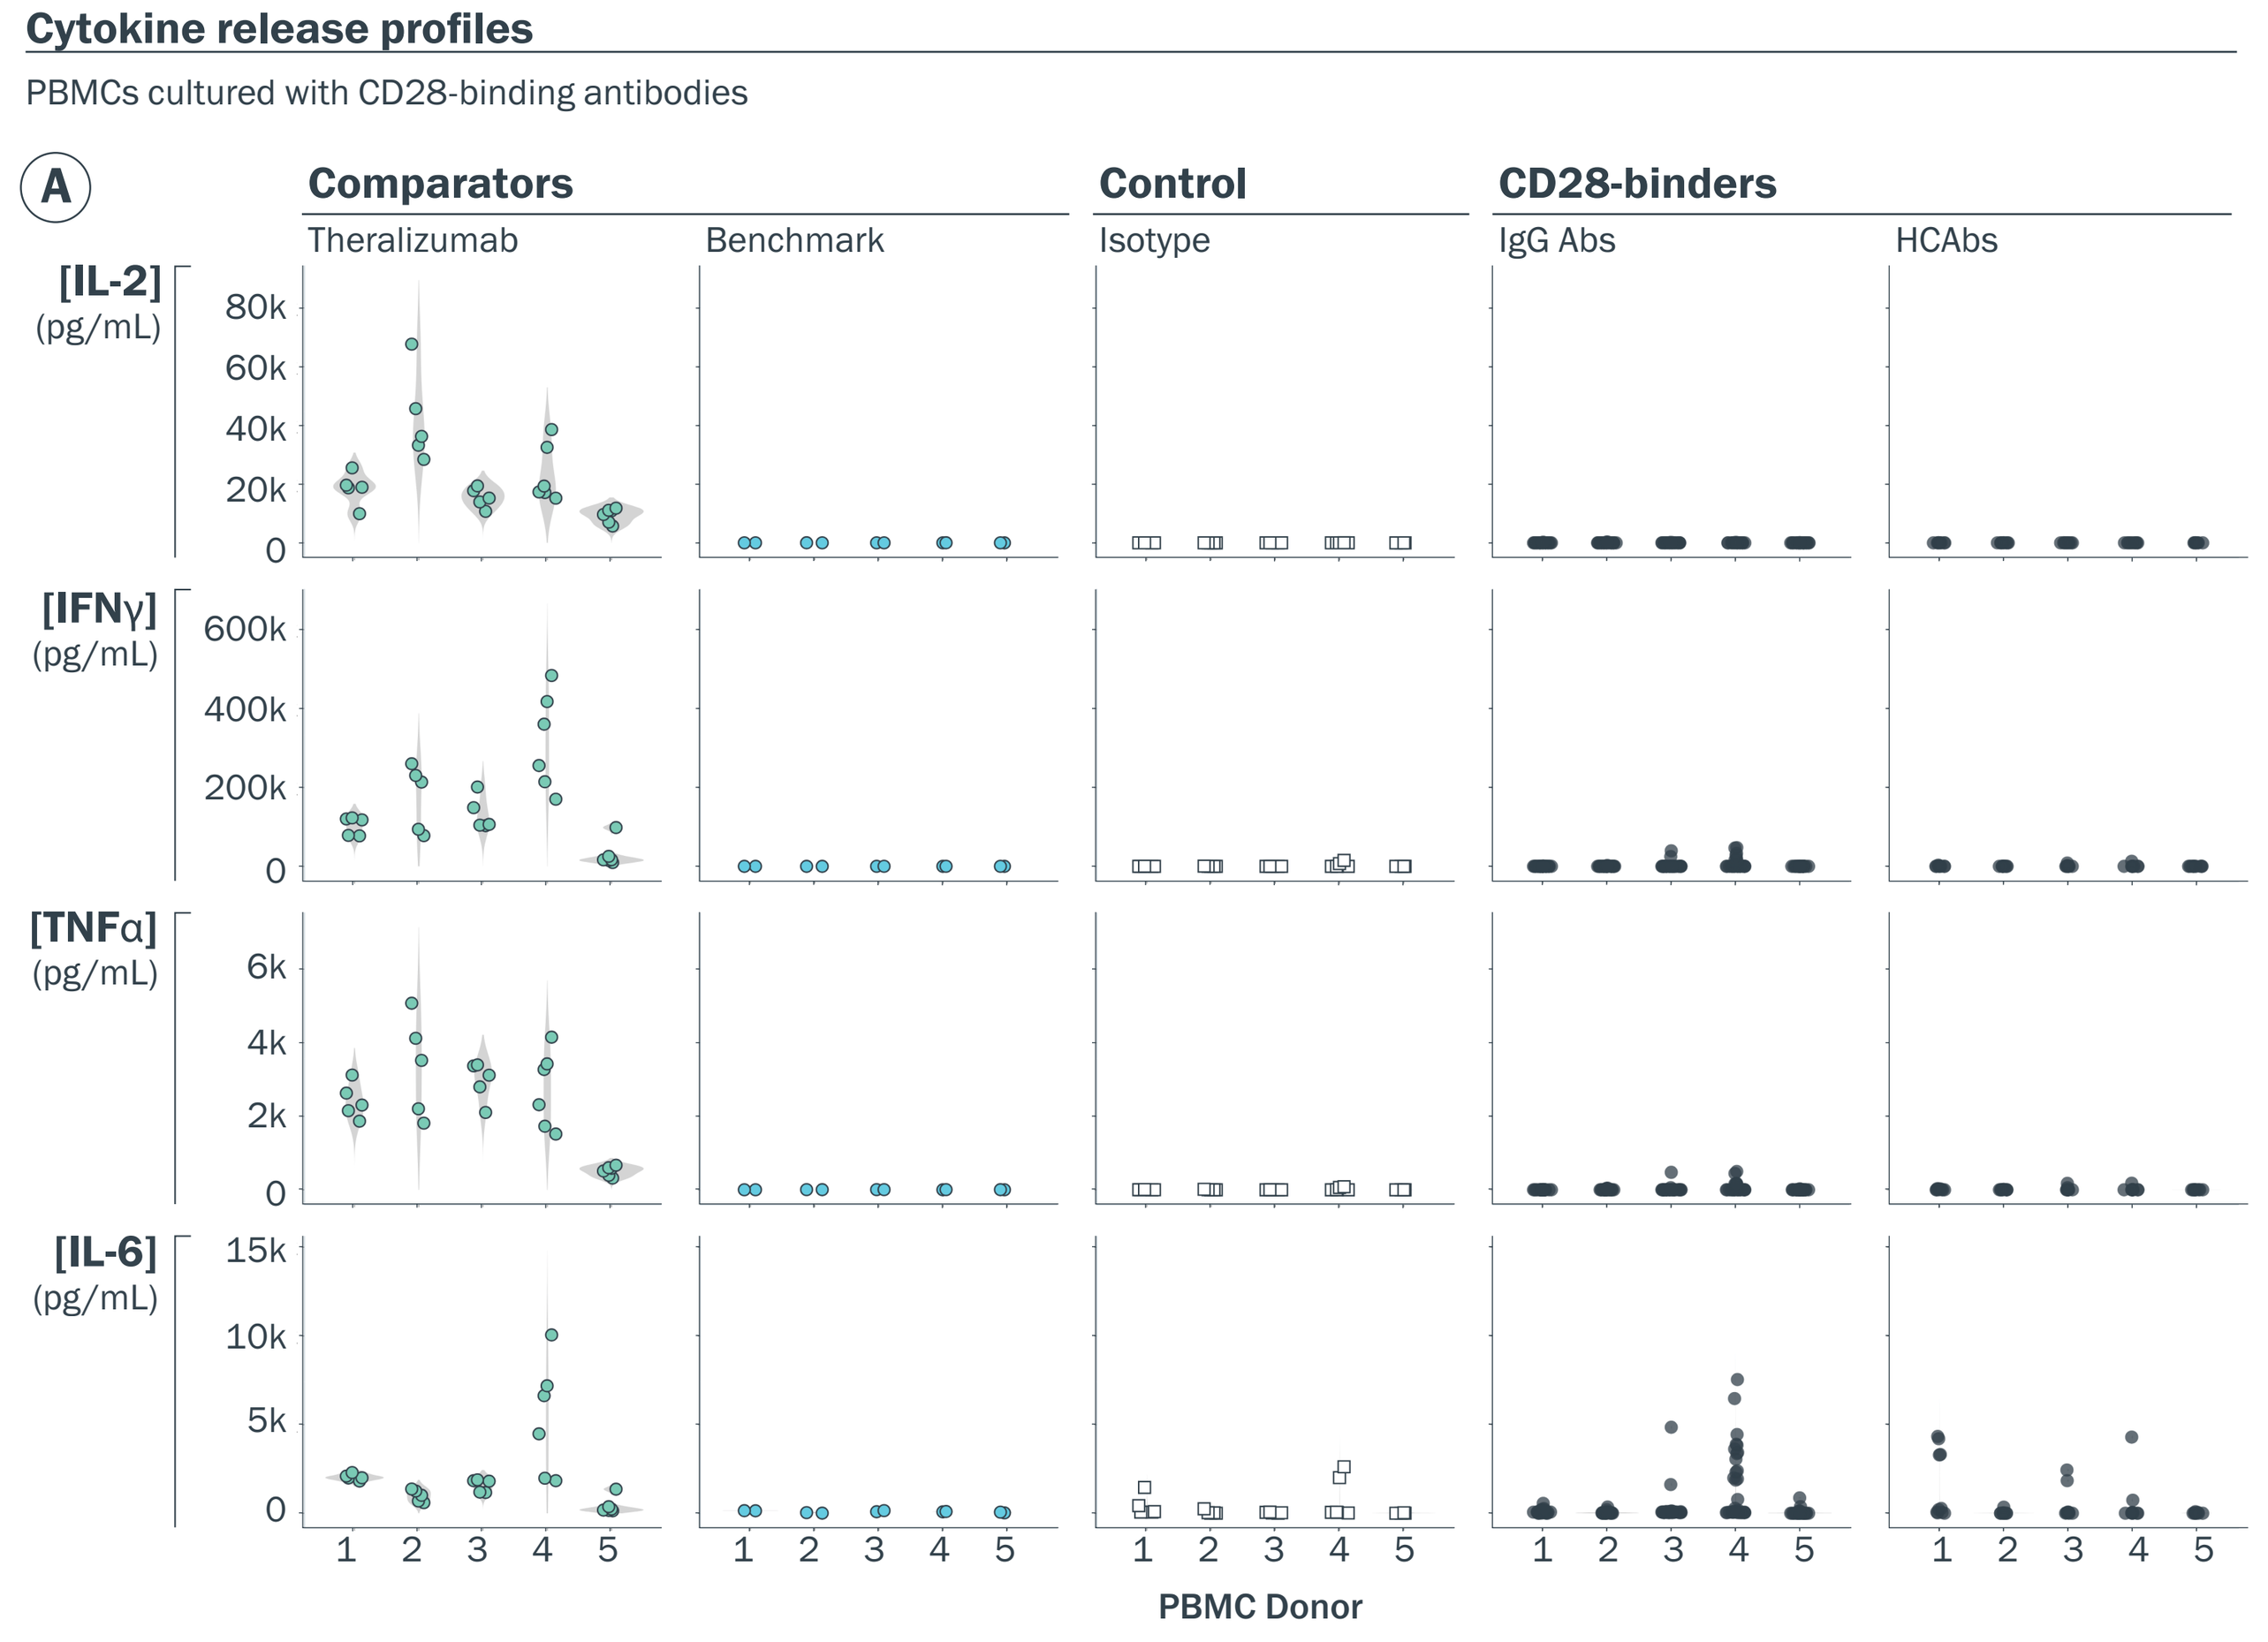
Cytokine release profiles
PBMCs cultured with CD28-binding antibodies
Comparators
Theralizumab
Benchmark
Control
Isotype
CD28-binders
IgG Abs
HCAbs
[IL-2] (pg/mL)
[IFNγ] (pg/mL)
[TNFα] (pg/mL)
[IL-6] (pg/mL)
PBMC Donor

Figure 3. CD28-binding antibodies do not show superagonist activity when mixed with peripheral blood mononuclear cells (PBMCs) from healthy donors. (A) Cytokine release profiles are shown for five PBMC donors that were cultured with wet-coated antibodies (five replicates/antibody, 22 total AbCellera CD28-binders), as previously described.[7] Values below the lower limit of detection are shown at y=0. Theralizumab is a known CD28 superagonist molecule. Antibodies were generated as a human IgG1 with a N297A mutation.

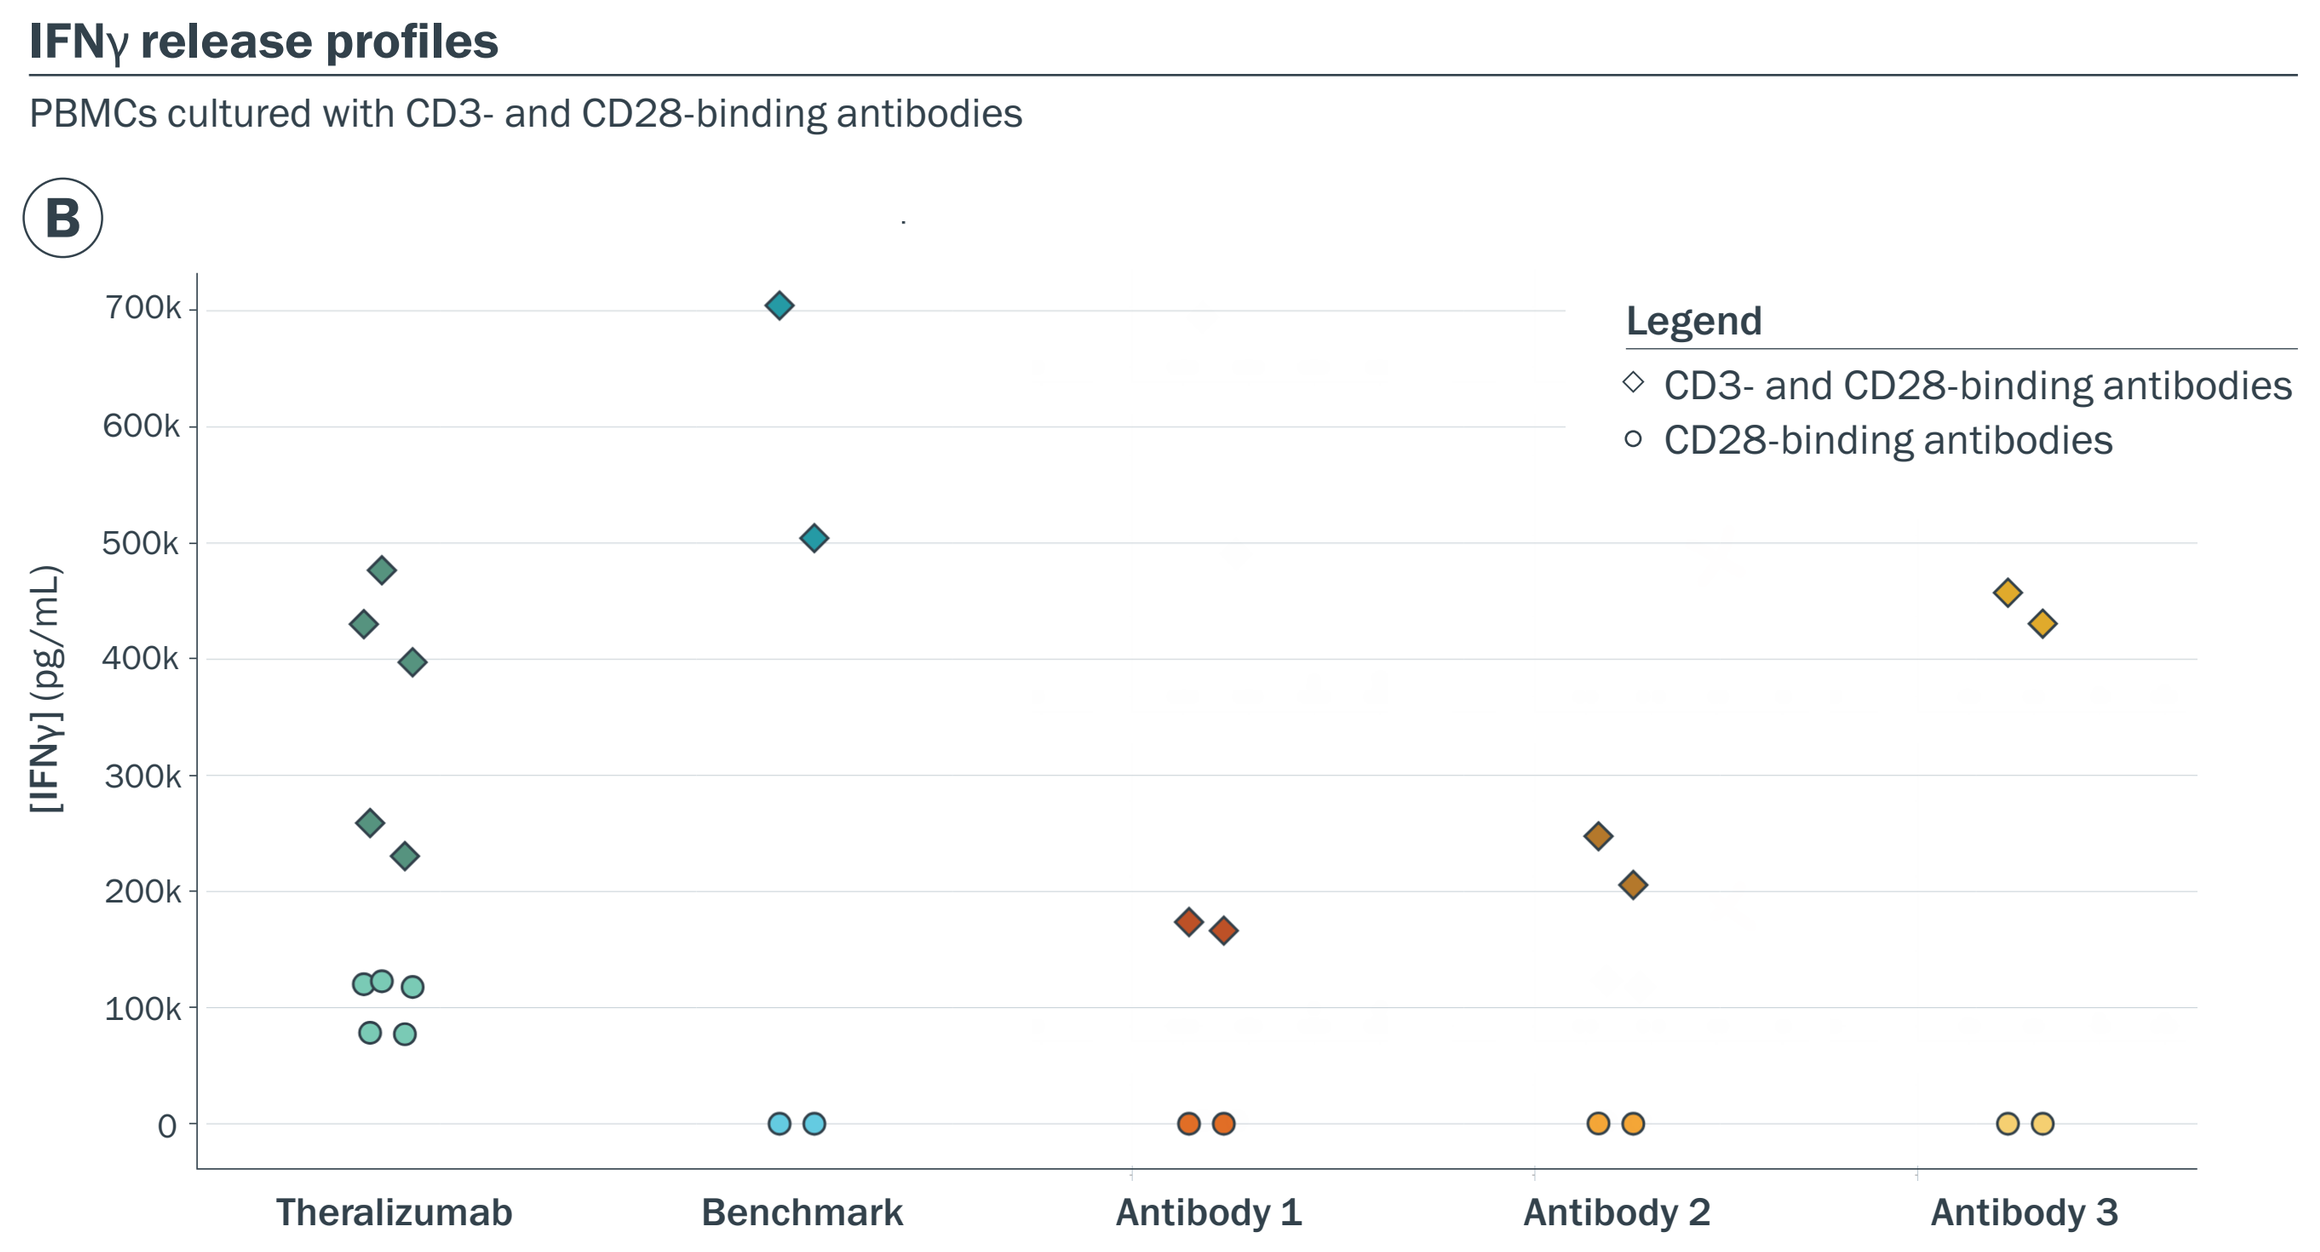
IFNγ release profiles
PBMCs cultured with CD3- and CD28-binding antibodies
[IFNγ] (pg/mL)
Legend
CD3- and CD28-binding antibodies
CD28-binding antibodies
Theralizumab
Benchmark
Antibody 1
Antibody 2
Antibody 3

(B) In a proof-of-concept study for MHC-independent costimulation (Fig. 1), PBMC donors were cultured with a combination of plate-bound CD28- and soluble CD3-binding (OKT3) antibodies. A single PBMC donor and representative cytokine, IFNγ, is shown. The costimulatory effect of CD28-binding antibodies was within the range of clinical benchmarks, as shown for three example antibodies.

Novel HCAb and IgG antibodies with diverse binding properties

479 HCAb and IgG antibodies with high sequence diversity

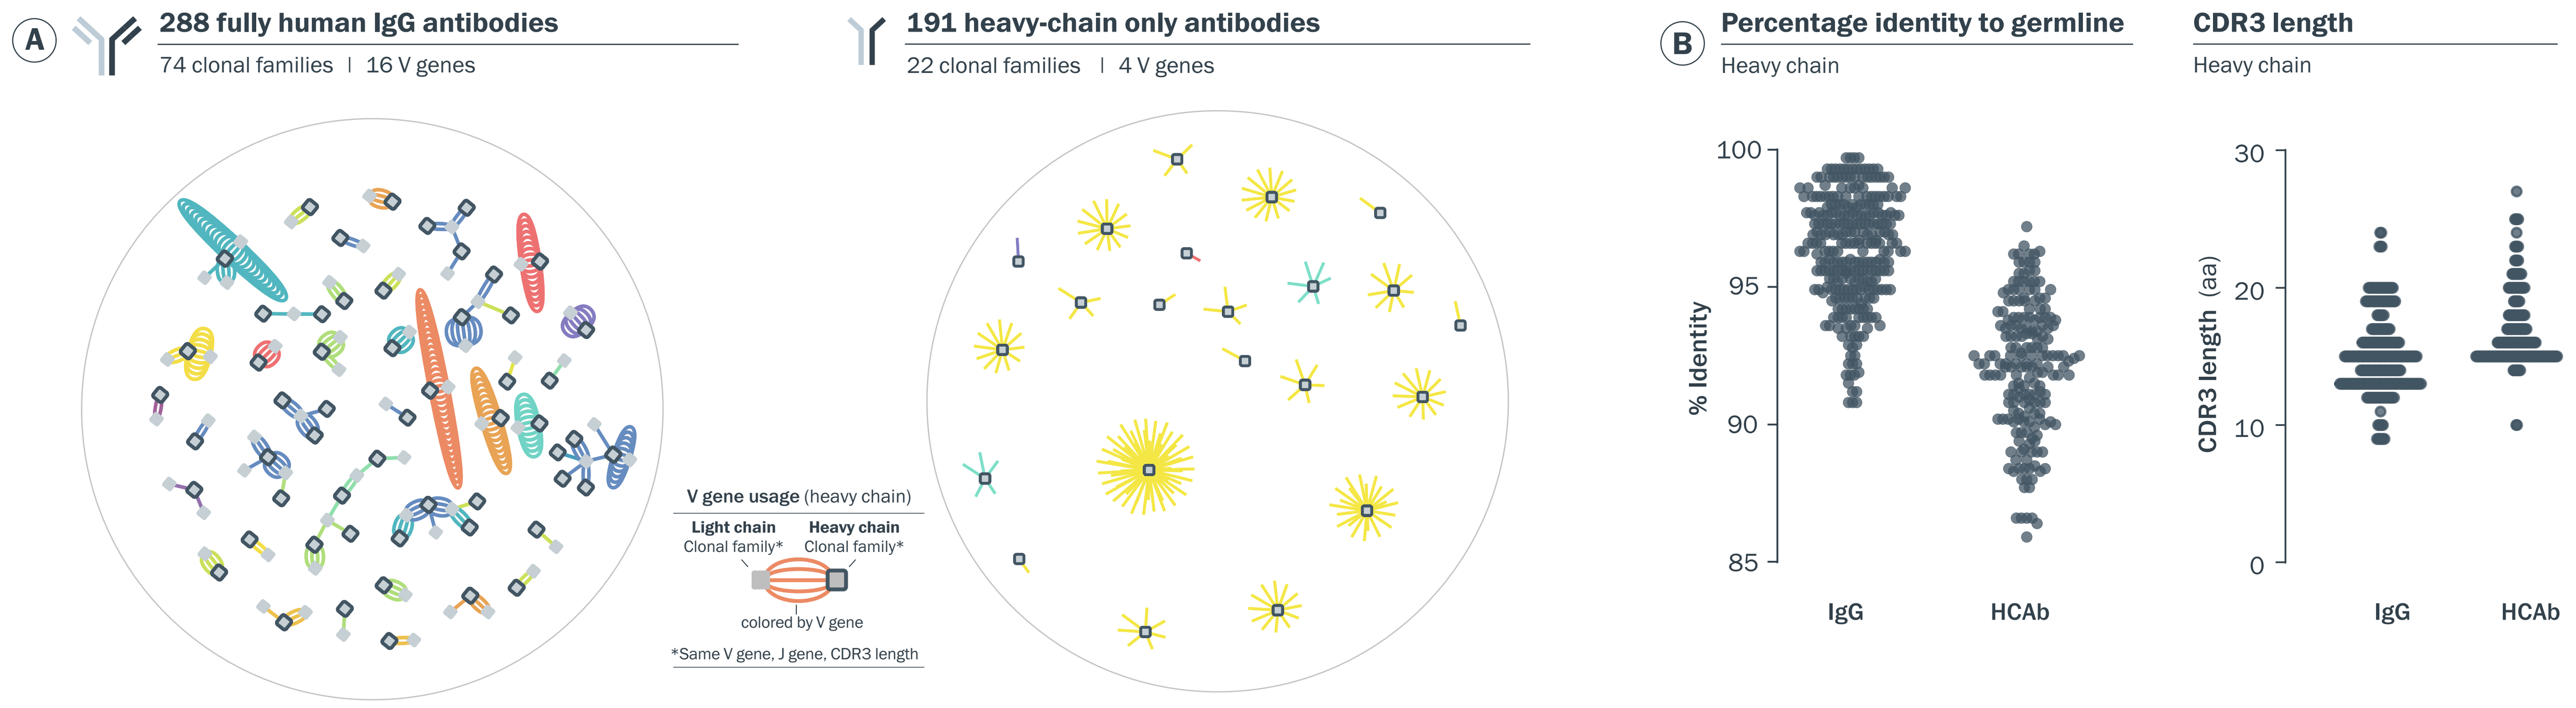
288 fully human IgG antibodies
74 clonal families | 16 V genes
191 heavy-chain only antibodies
22 clonal families | 4 V genes
V gene usage (heavy chain)
Light chain Clonal family*
Heavy chain Clonal family*
colored by V gene
*Same V gene, J gene, CDR3 length
Percentage identity to germline
Heavy chain
% Identity
CDR3 length
Heavy chain
CDR3 length (aa)
IgG
HCAb

Figure 4. 479 CD28-specific HCAb and IgG antibodies were identified from immunization and deep single-cell screening. (A) To discover the functional antibodies in the above figures, we immunized humanized mice and camelids to identify IgG and HCAbs, respectively, that bind to both human and cyno CD28. Antibody sequence diversity was visualized using Celium™. **(B)** Antibodies showed a broad range of percentage identity to germline and CDR3 length, highlighting diversity in sequences obtained.

CD28-binding antibodies with epitopic diversity and a range of avidities

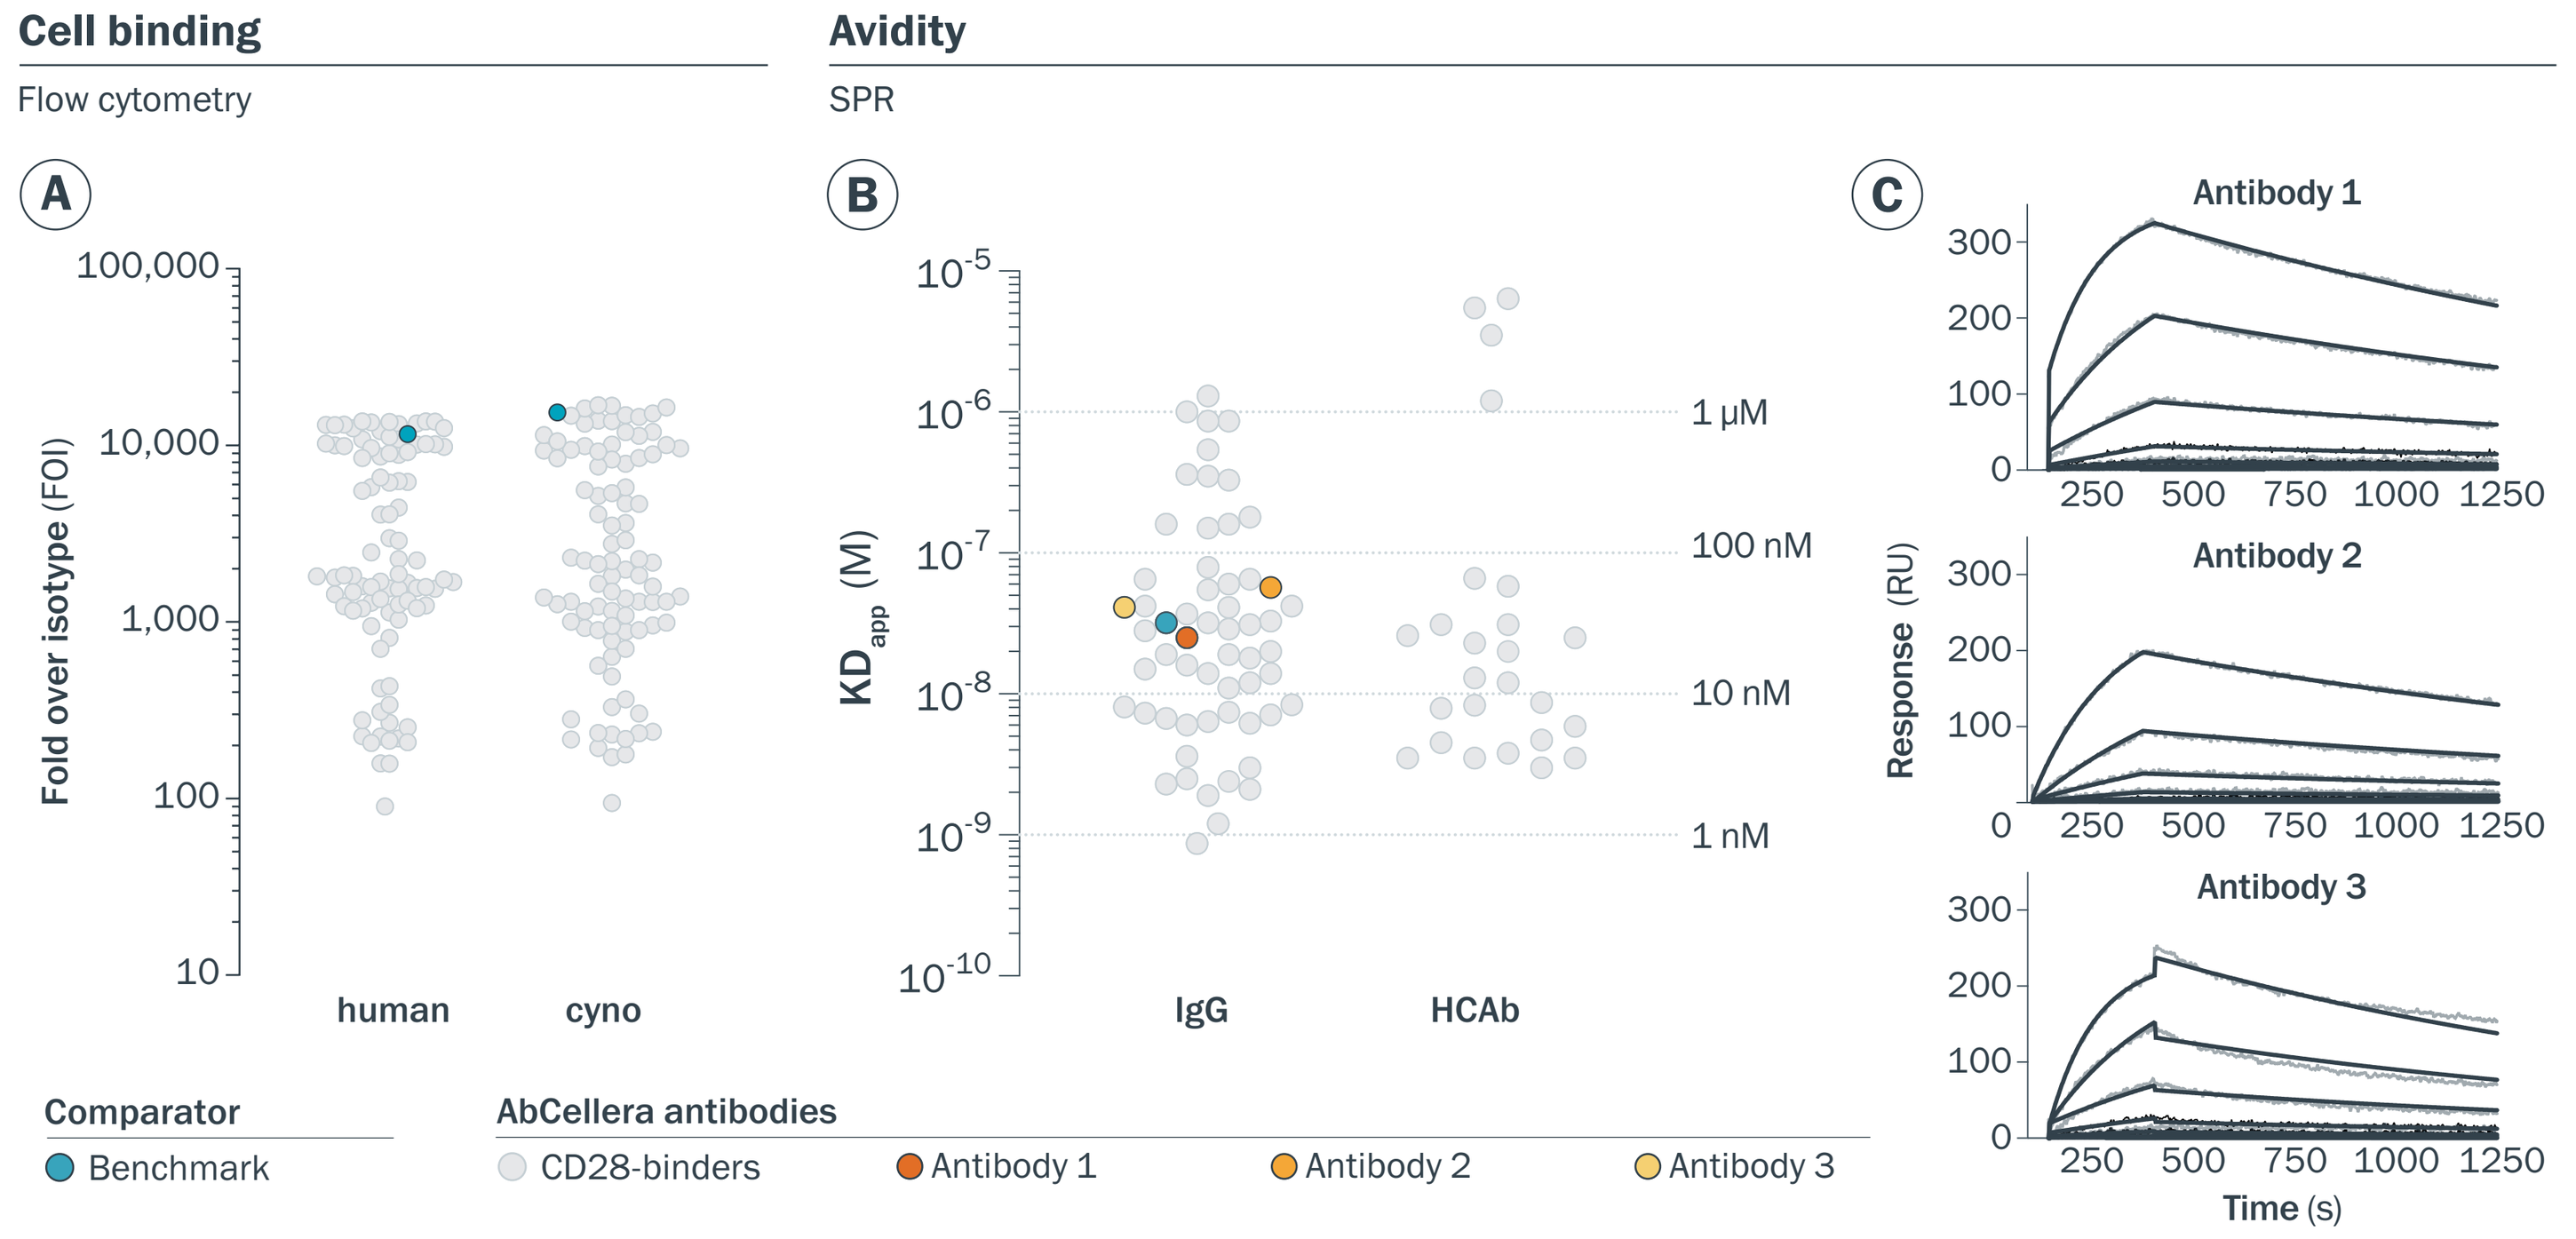
Cell binding
Flow cytometry
Fold over isotype (FOI)
human
cyno
Avidity
SPR
KDapp (M)
1 μM
100 nM
10 nM
1 nM
IgG
HCAb
Antibody 1
Antibody 2
Antibody 3
Response (RU)
Time (s)
Comparator
Benchmark
AbCellera antibodies
CD28-binders
Antibody 1
Antibody 2
Antibody 3

Figure 5. Antibodies showed diversity in epitopes and binding avidities. (A) A subset of cross-reactive antibodies (65 IgGs and 27 HCAbs, at 25 nM) were selected for expression and assessed for binding to human and cyno CD28-expressing cells. The median fluorescence intensity for each antibody was normalized to the isotype control, with positive binding (fold over isotype ⩾ 10) confirmed in 90 of the 92 antibodies. **(B)** Avidity was measured using the CD28 extracellular domain by surface plasmon resonance. Antibodies showed diverse binding avidities, ranging from 870 pM to 6.40 μM. **(C)** Sensorgrams show three example IgG antibodies (1-3) with diversity in on and off rate.

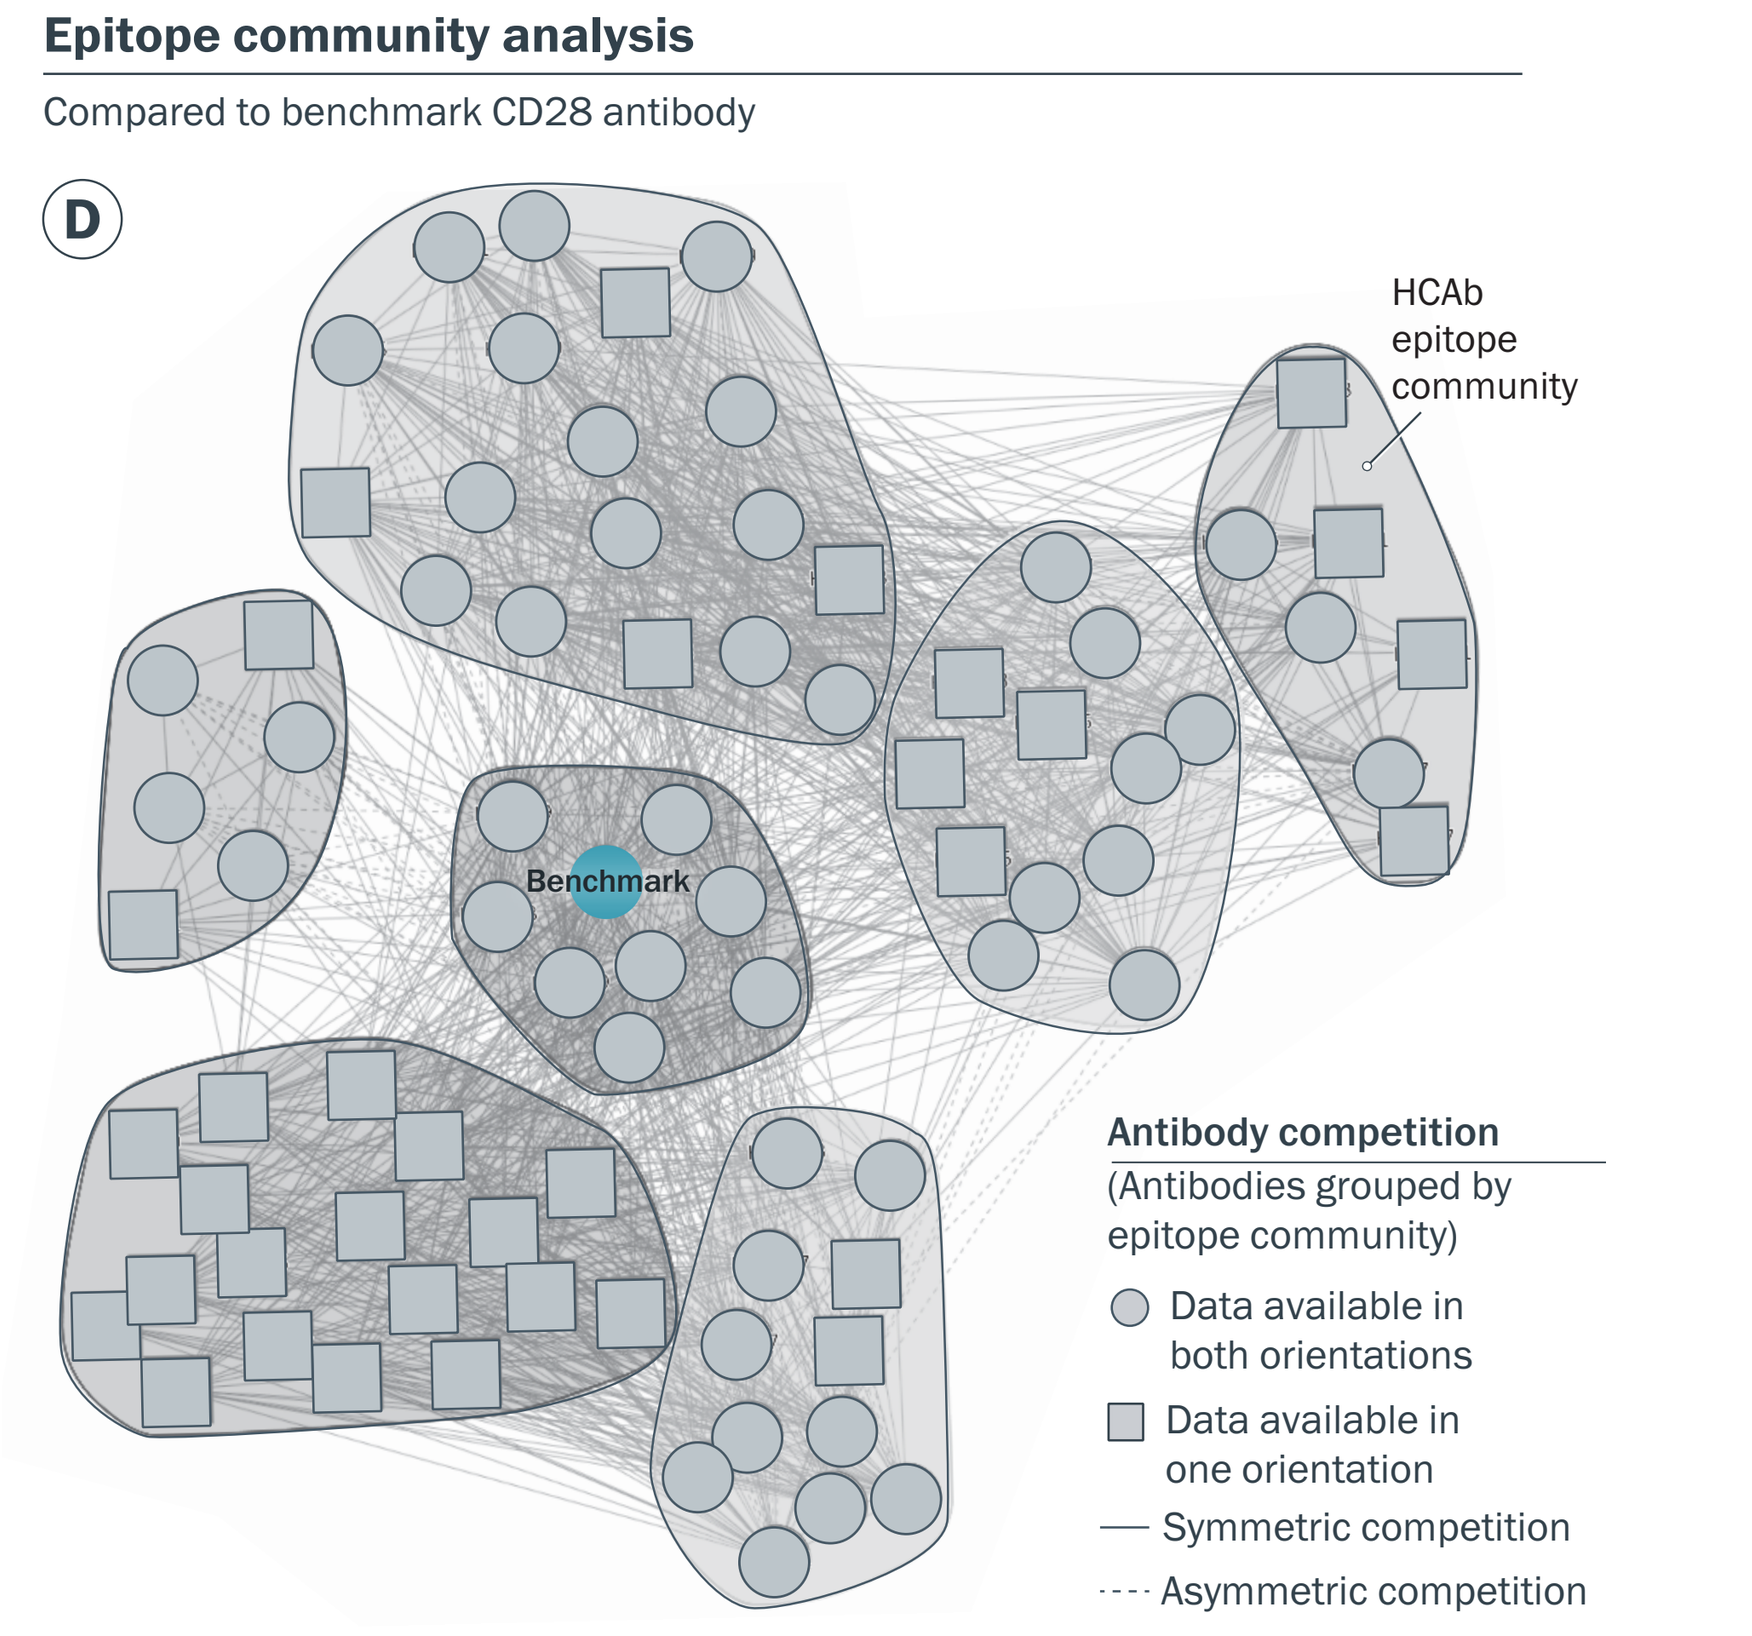
Epitope community analysis
Compared to benchmark CD28 antibody
HCAb epitope community
Benchmark
Antibody competition
(Antibodies grouped by epitope community)
Data available in both orientations
Data available in one orientation
Symmetric competition
Asymmetric competition

(D) Epitope binning was performed, with communities determined by ward.D hierarchical clustering based on competition events. 81 antibodies clustered in seven epitope communities and the majority of antibodies compete with the benchmark for binding to CD28, consistent with the limited extracellular space.

Biophysical characterization shows antibodies are of high quality

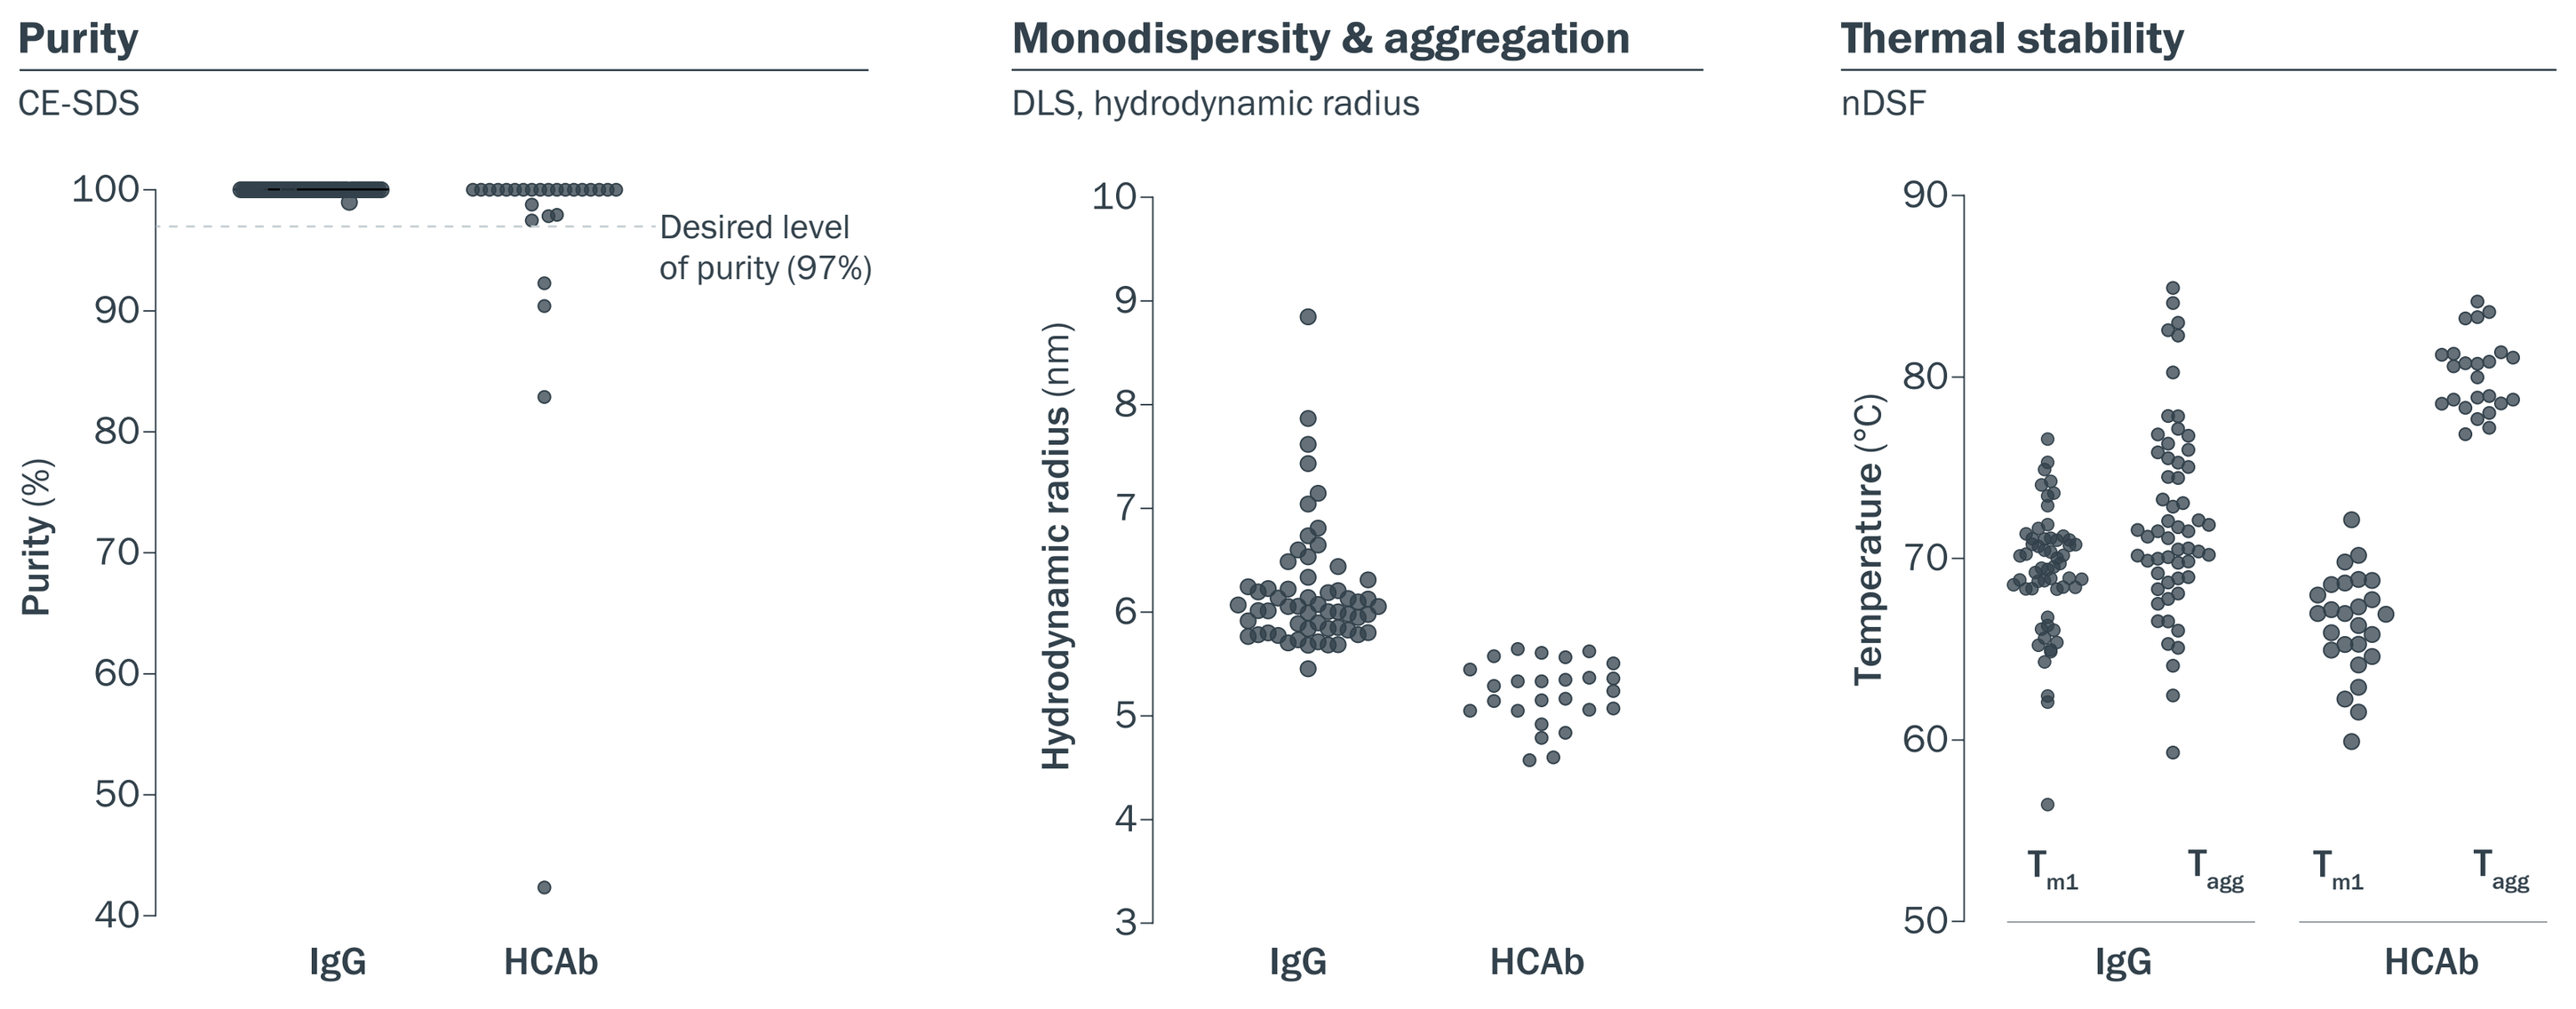
Purity
CE-SDS
Purity (%)
Desired level of purity (97%)
Monodispersity & aggregation
DLS, hydrodynamic radius
Hydrodynamic radius (nm)
Thermal stability
nDSF
Temperature (°C)
Tm1
Tagg
IgG
HCAb

Figure 6. Preliminary analytical and biophysical characterization reveals favorable early developability properties. The majority of antibodies showed a high level of purity (> 97%), a hydrodynamic radius within the expected range (IgGs: 5.3-6.5 nm; HCAbs: 4.7-6.3 nm), and typical thermal stability profiles.